Safety and efficacy of maralixibat in patients with primary sclerosing cholangitis: an open-label proof-of-concept study

Christopher L. Bowlus,[1] Bertus Eksteen,[2] Angela Cheung,[3] Douglas Thorburn,[4] Cynthia A. Moylan,[5] Paul J. Pockros,[6] Lisa M. Forman,[7] Alejandro Dorenbaum,[8] Gideon M. Hirschfield,[9] Ciara Kennedy,[10] Joan Gu,[11] George Apostol,[12] Pamela Vig,[13] Cynthia Levy[14]

[1]Division of Gastroenterology and Hepatology, University of California Davis School of Medicine, Sacramento, CA, USA; [2]University of Calgary, Aspen Woods Clinic, Calgary, AB, Canada; [3]Division of Gastroenterology and Hepatology, Mayo Clinic, Rochester, MN, USA; [4]Sheila Sherlock Liver Centre, Royal Free Hospital and Institute of Liver and Digestive Health, University College London, Royal Free Campus, Hampstead, London, UK; [5]Division of Gastroenterology and Hepatology, Duke University, Durham, NC, USA; [6]Scripps Clinic and Scripps Translational Science Institute, La Jolla, CA, USA; [7]Division of Gastroenterology-Hepatology, University of Colorado, Aurora, CO, USA; [8]Stanford University, Palo Alto, CA, USA; [9]Toronto Centre for Liver Disease, University Health Network, University of Toronto, Toronto, ON, Canada; [10]Amplyx Pharmaceuticals, San Diego, CA, USA; [11]Takeda Pharmaceuticals, Lexington, MA, USA; [12]Takeda Pharmaceuticals, Zug, Switzerland; [13]Mirum Pharmaceuticals, Foster City, CA, USA; [14]Division of Hepatology, University of Miami Miller School of Medicine, Miami, FL, USA

1262

Background

- Primary sclerosing cholangitis (PSC) is a chronic liver disease characterized by progressive cholestasis.
 - Cholestasis can often lead to severe pruritus, which significantly impairs quality of life.
- Maralixibat (formerly SHP625, LUM001) is a minimally absorbed, selective inhibitor of the apical sodium-dependent bile acid transporter (ASBT).
 - Maralixibat interrupts enterohepatic circulation of bile acids.[1]
- Maralixibat may control pruritus in patients with PSC.
 - ASBT inhibition with maralixibat has been shown to reduce serum bile acid (sBA) levels and reduce pruritus in other cholestatic liver diseases.[2,3]

Objectives

- To evaluate the safety and tolerability of maralixibat at doses of up to 10 mg/day for 14 weeks in adults with PSC.
- To assess changes from baseline in sBA levels, pruritus, and biomarkers of cholestasis, liver function and PSC.

Methods

Design

- CAMEO (ClinicalTrials.gov: NCT02061540) was a 14-week, single-arm, open-label, phase 2a, proof-of-concept study of maralixibat.
- A 6-week dose-escalation period (maralixibat 0.5 mg, 1 mg, 2.5 mg, 5 mg and 7.5 mg/day) was followed by an 8-week dose-maintenance period (maralixibat 10 mg/day) and a 4-week follow-up period.

Participants

- Eligible patients were adults aged 18–80 years with a diagnosis of PSC, defined as: history of alkaline phosphatase (ALP) levels > 1.5 × ULN, biliary obstruction and histological findings consistent with PSC (if previously biopsied).
- Pruritus was not required for inclusion.

Study Assessments

- Treatment-emergent adverse events (TEAEs) were assessed regularly.
- Pruritus was assessed using Adult Itch Reported Outcome (ItchRO) weekly sum scores and average daily scores (mean score over a 7-day period).
 - Participants self-reported the ItchRO scores daily in an e-diary on a scale of 0–10 (0, no pruritus; 10, most severe pruritus).
- Other efficacy assessments included change from baseline in levels of sBA, 7-α-hydroxy-cholesten-3-one (7αC4, a marker of *de novo* bile acid synthesis), autotaxin (a marker of cholestatic pruritus[4]), low-density lipoprotein cholesterol (LDL-C), total cholesterol and liver enzymes.
- Outcomes were assessed overall and in subgroups comprising patients with any pruritus at baseline (ItchRO daily score > 0) and those with an ItchRO average daily score ≥ 4 out of 10 at baseline.
- Changes from baseline to week 14 or early termination (ET) were analyzed using paired *t*-tests or Wilcoxon signed-rank tests.

Results

Disposition and Demographics

- Of 27 enrolled participants, 23 (85.2%) completed the study; demographic and baseline characteristics are shown in **Table 1**.
- Six participants did not receive the highest dose (10 mg) due to ET (n = 1) or down-titration for tolerability (n = 5).
 - Doses were 1 mg (n = 1), 2.5 mg (n = 2), 5 mg (n = 2) and 7.5 mg (n = 1).

Pruritus, sBA and 7αC4

- Mean ItchRO weekly sum scores decreased significantly from baseline overall, and in the pruritus subgroups (**Figure 1**).
- The six participants with baseline ItchRO ≥ 4 all had improvements in pruritus (**Figure 2**).
- Mean sBA levels decreased and mean 7αC4 levels increased from baseline overall and in participants with an ItchRO daily score ≥ 4 at baseline (**Figure 3**).

Additional Outcomes

- Mean autotaxin and LDL-C levels decreased from baseline overall and in participants with an ItchRO daily score ≥ 4 at baseline (**Figure 4**).
- Total cholesterol levels decreased and conjugated bilirubin levels increased significantly from baseline; changes in total bilirubin, alanine aminotransferase, aspartate aminotransferase and ALP levels were not statistically significant (**Table 2**).

Table 1. Baseline demographic characteristics

		Overall sample (N = 27)
Male sex, n (%)		18 (66.7)
Mean age, years (SD)		43.7 (11.35)
Race, n (%)	White	23 (85.2)
	Other[a]	4 (14.8)
Symptoms of PSC, n (%)	Ulcerative colitis	15 (55.6)
	Inflammatory bowel disease	12 (44.4)
Mean alkaline phosphatase, U/L (SD)		472 (316.9)
Mean aspartate aminotransferase, U/L (SD)		88 (43.7)
Mean alanine aminotransferase, U/L (SD)		109 (79.0)

[a]Asian (n = 1, 3.7%), Black (n = 1, 3.7%), multiple (n = 2, 7.4%).
PSC, primary sclerosing cholangitis; SD, standard deviation.

Figure 1. ItchRO weekly sum score

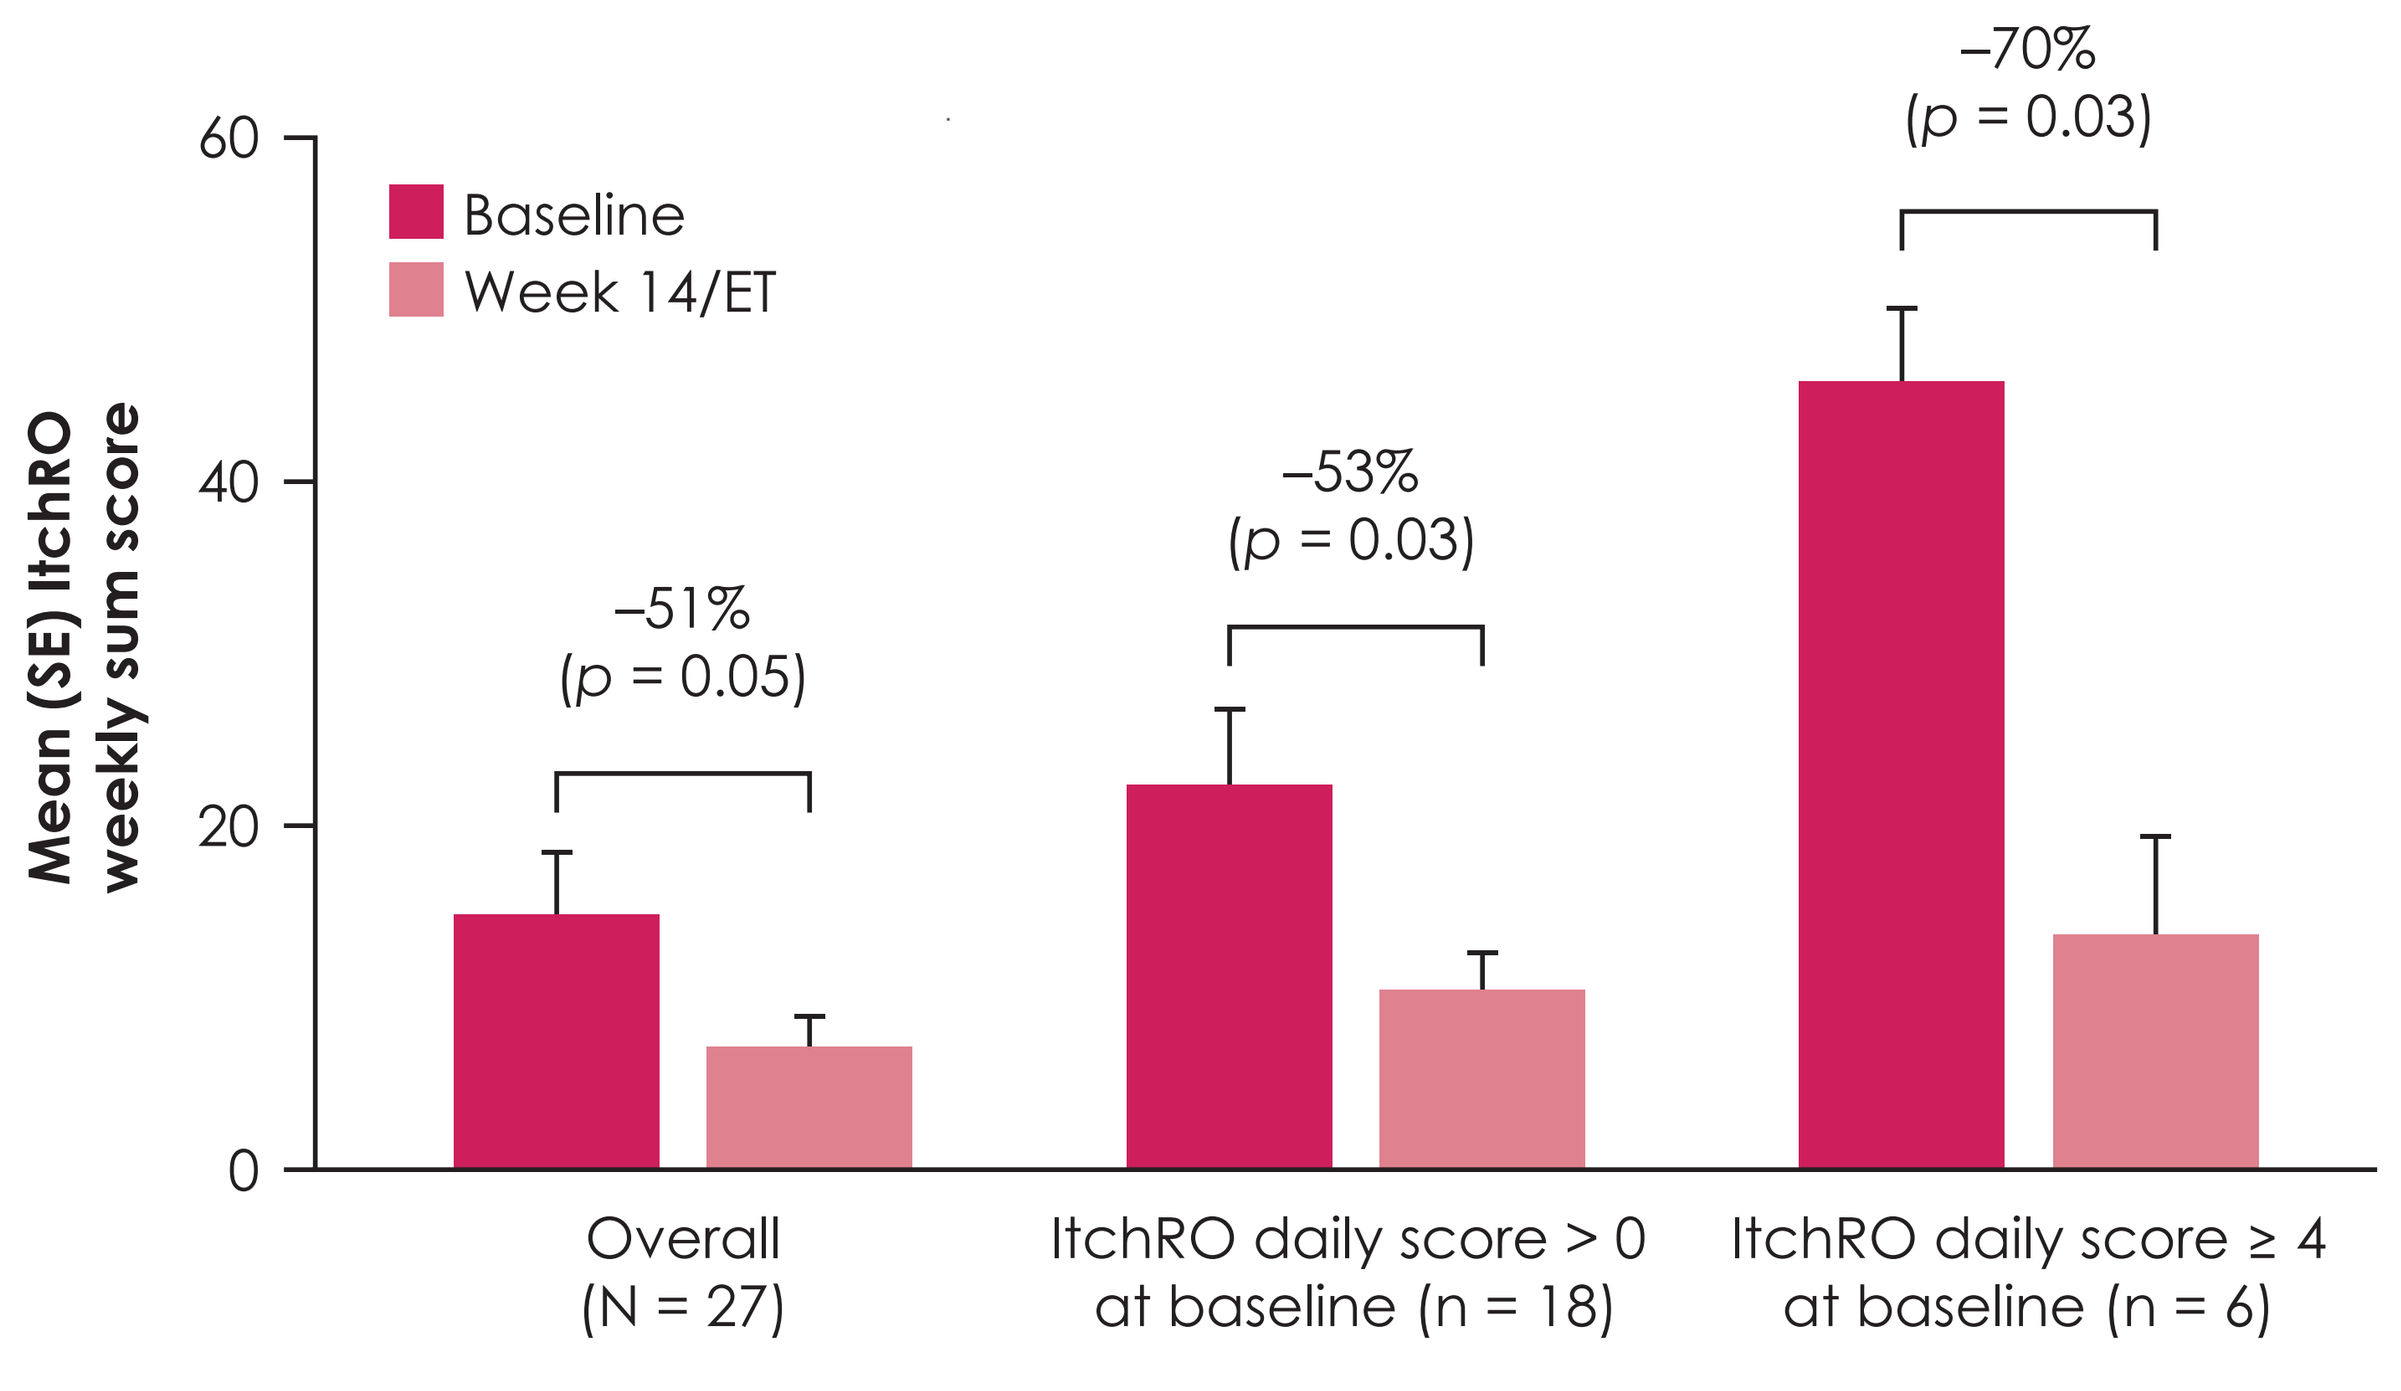
Baseline
Week 14/ET
Mean (SE) ItchRO weekly sum score
60
40
20
0
–51%
(p = 0.05)
–53%
(p = 0.03)
–70%
(p = 0.03)
Overall
(N = 27)
ItchRO daily score > 0
at baseline (n = 18)
ItchRO daily score ≥ 4
at baseline (n = 6)

Early termination was the last post-baseline value before or within 7 days of the last dose.
ET, early termination; ItchRO, Adult Itch Reported Outcome (0–10 scale); SE, standard error.

Figure 2. ItchRO scores in the six participants with an ItchRO daily score ≥ 4 at baseline

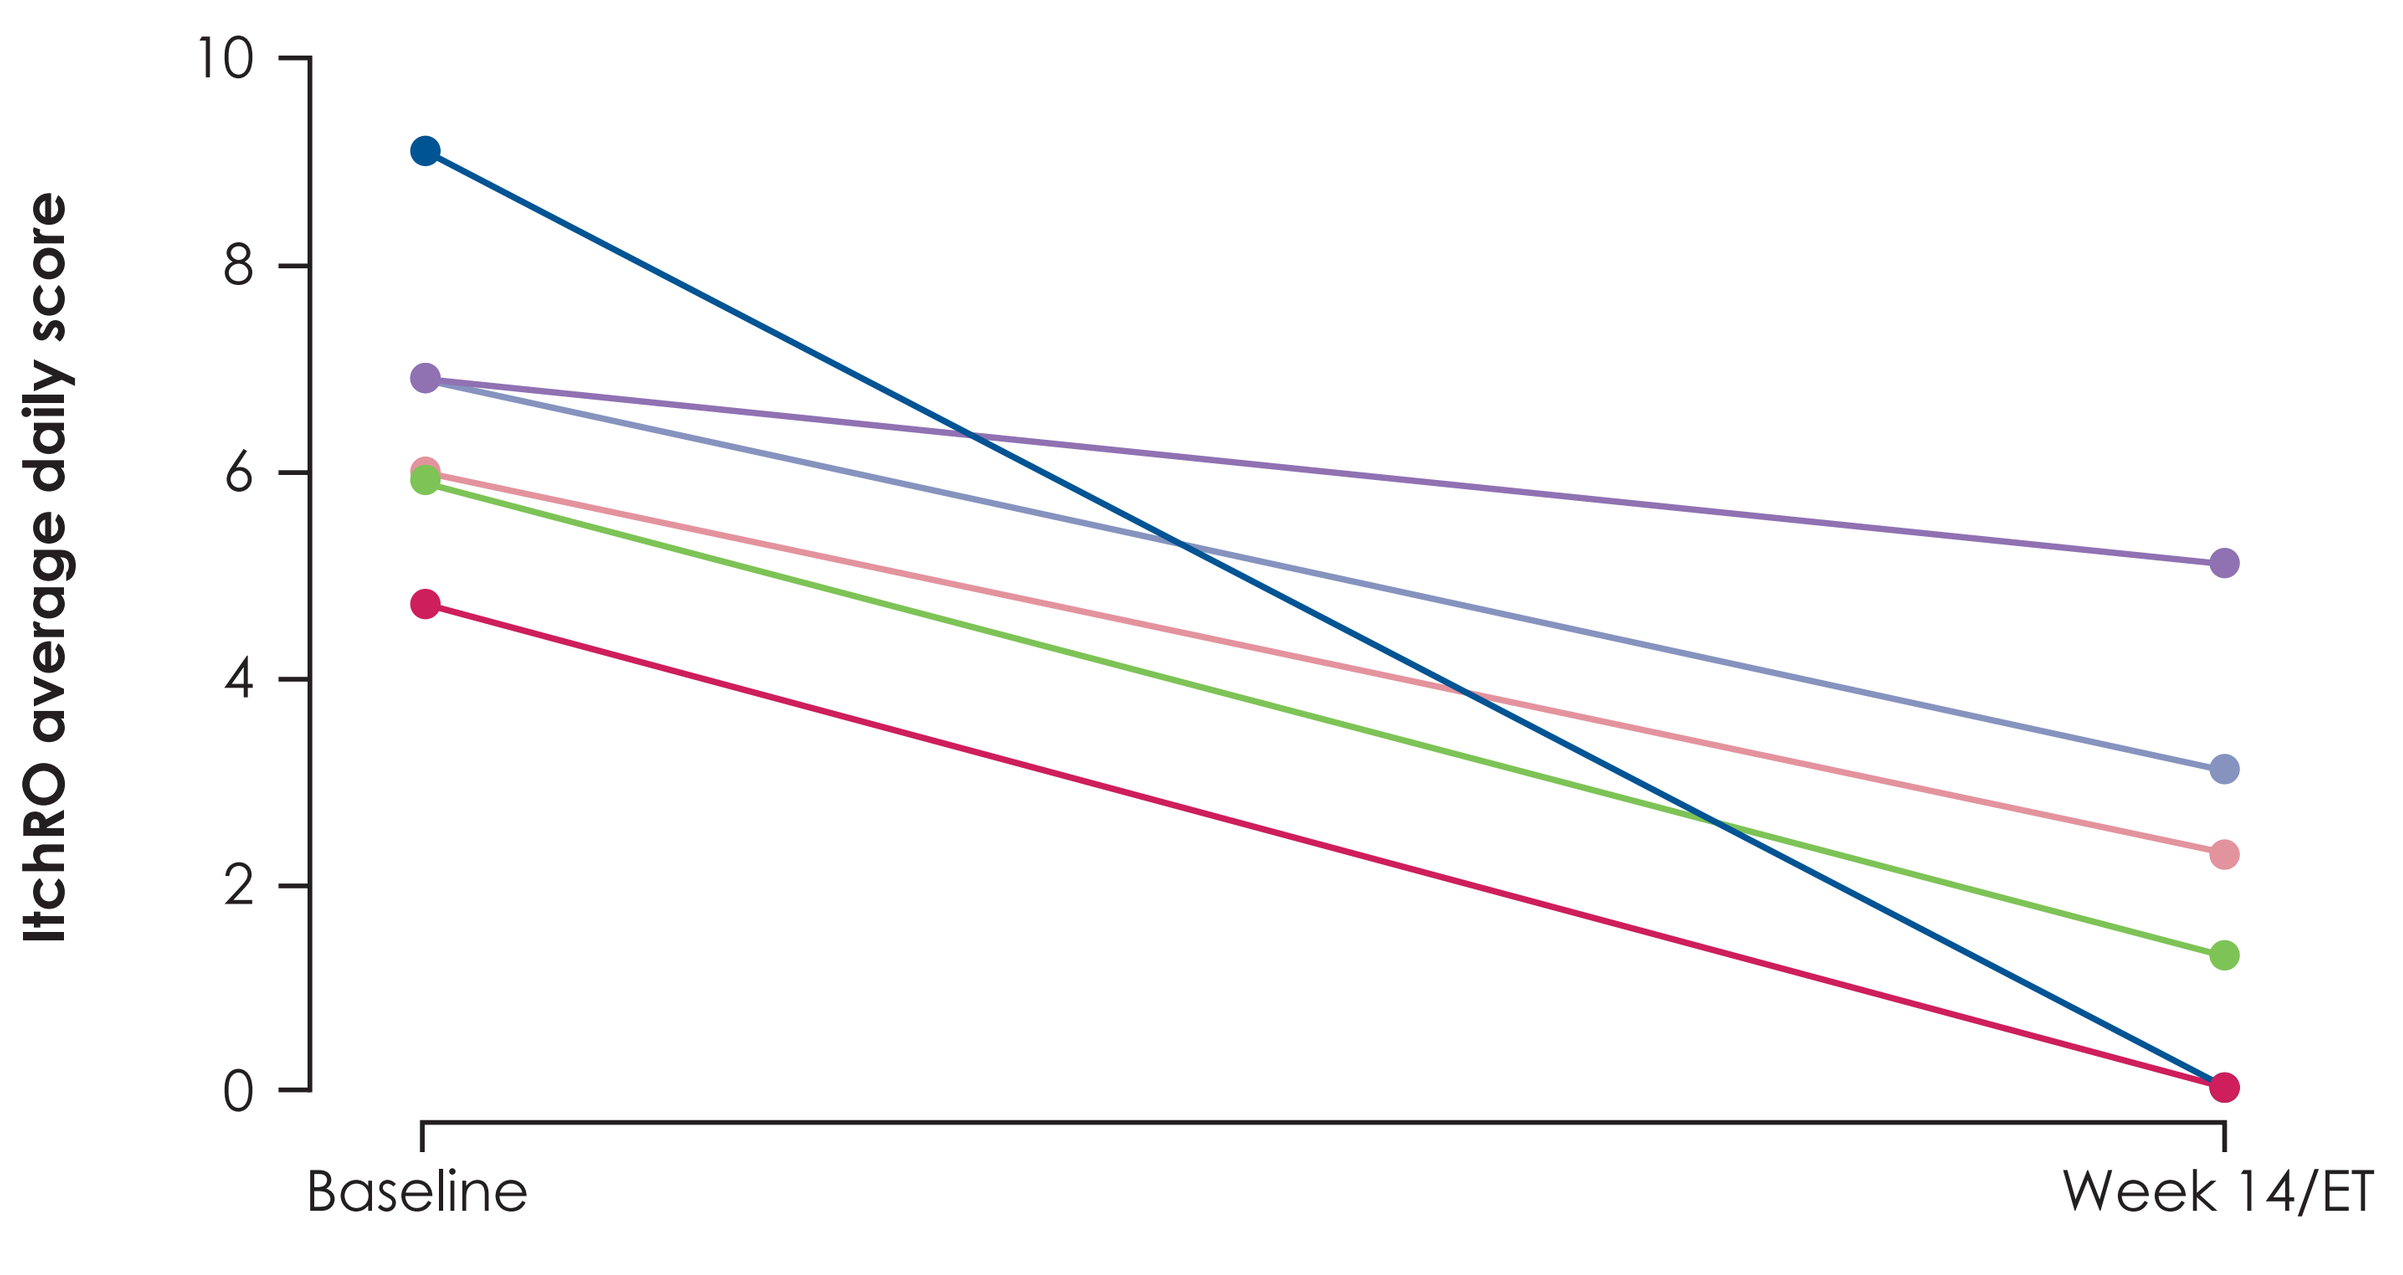
ItchRO average daily score
10
8
6
4
2
0
Baseline
Week 14/ET

Early termination was the last post-baseline value before or within 7 days of the last dose.
ET, early termination; ItchRO, Adult Itch Reported Outcome (0–10 scale).

Figure 3. Serum bile acid and 7-α-hydroxy-cholesten-3-one levels

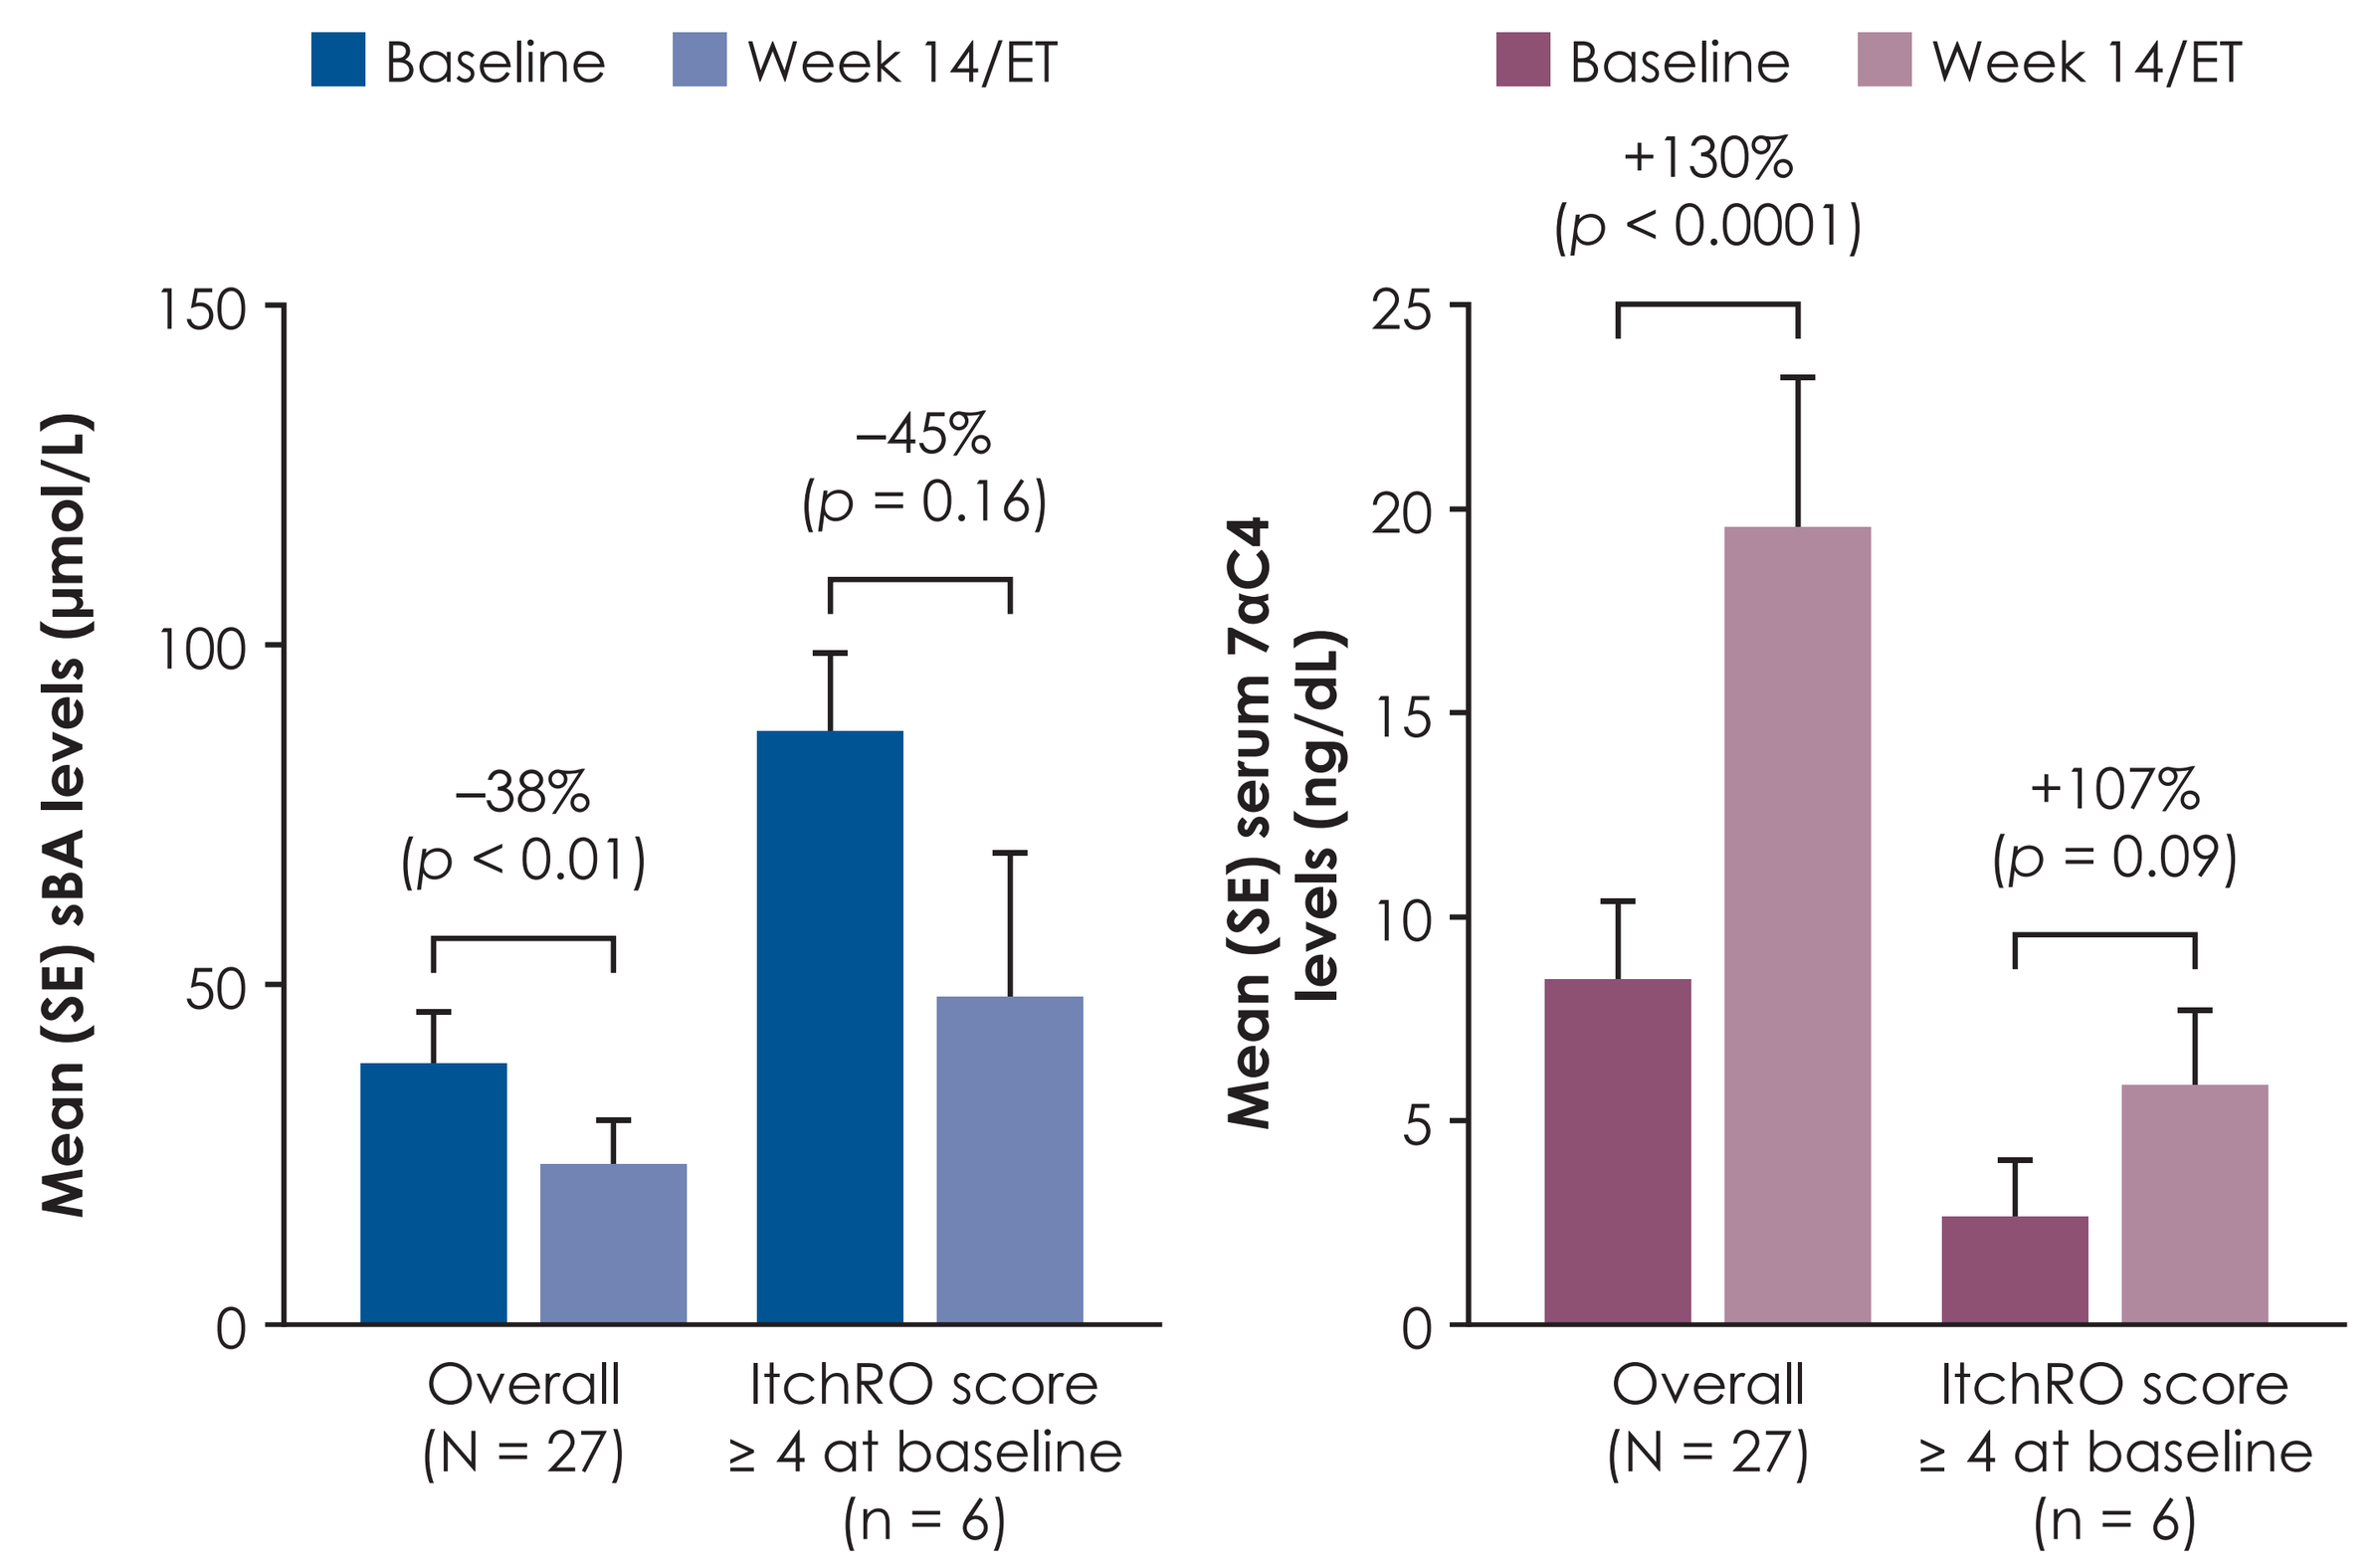
Baseline
Week 14/ET
Mean (SE) sBA levels (µmol/L)
150
100
50
0
–38%
(p < 0.01)
–45%
(p = 0.16)
Overall
(N = 27)
ItchRO score
≥ 4 at baseline
(n = 6)
Baseline
Week 14/ET
Mean (SE) serum 7αC4 levels (ng/dL)
25
20
15
10
5
0
+130%
(p < 0.0001)
+107%
(p = 0.09)
Overall
(N = 27)
ItchRO score
≥ 4 at baseline
(n = 6)

Early termination was the last post-baseline value before or within 7 days of the last dose.
7αC4, 7-α-hydroxy-cholesten-3-one; ET, early termination; ItchRO, Adult Itch Reported Outcome (0–10 scale); sBA, serum bile acid; SE, standard error.

Figure 4. Autotaxin and low-density lipoprotein cholesterol levels

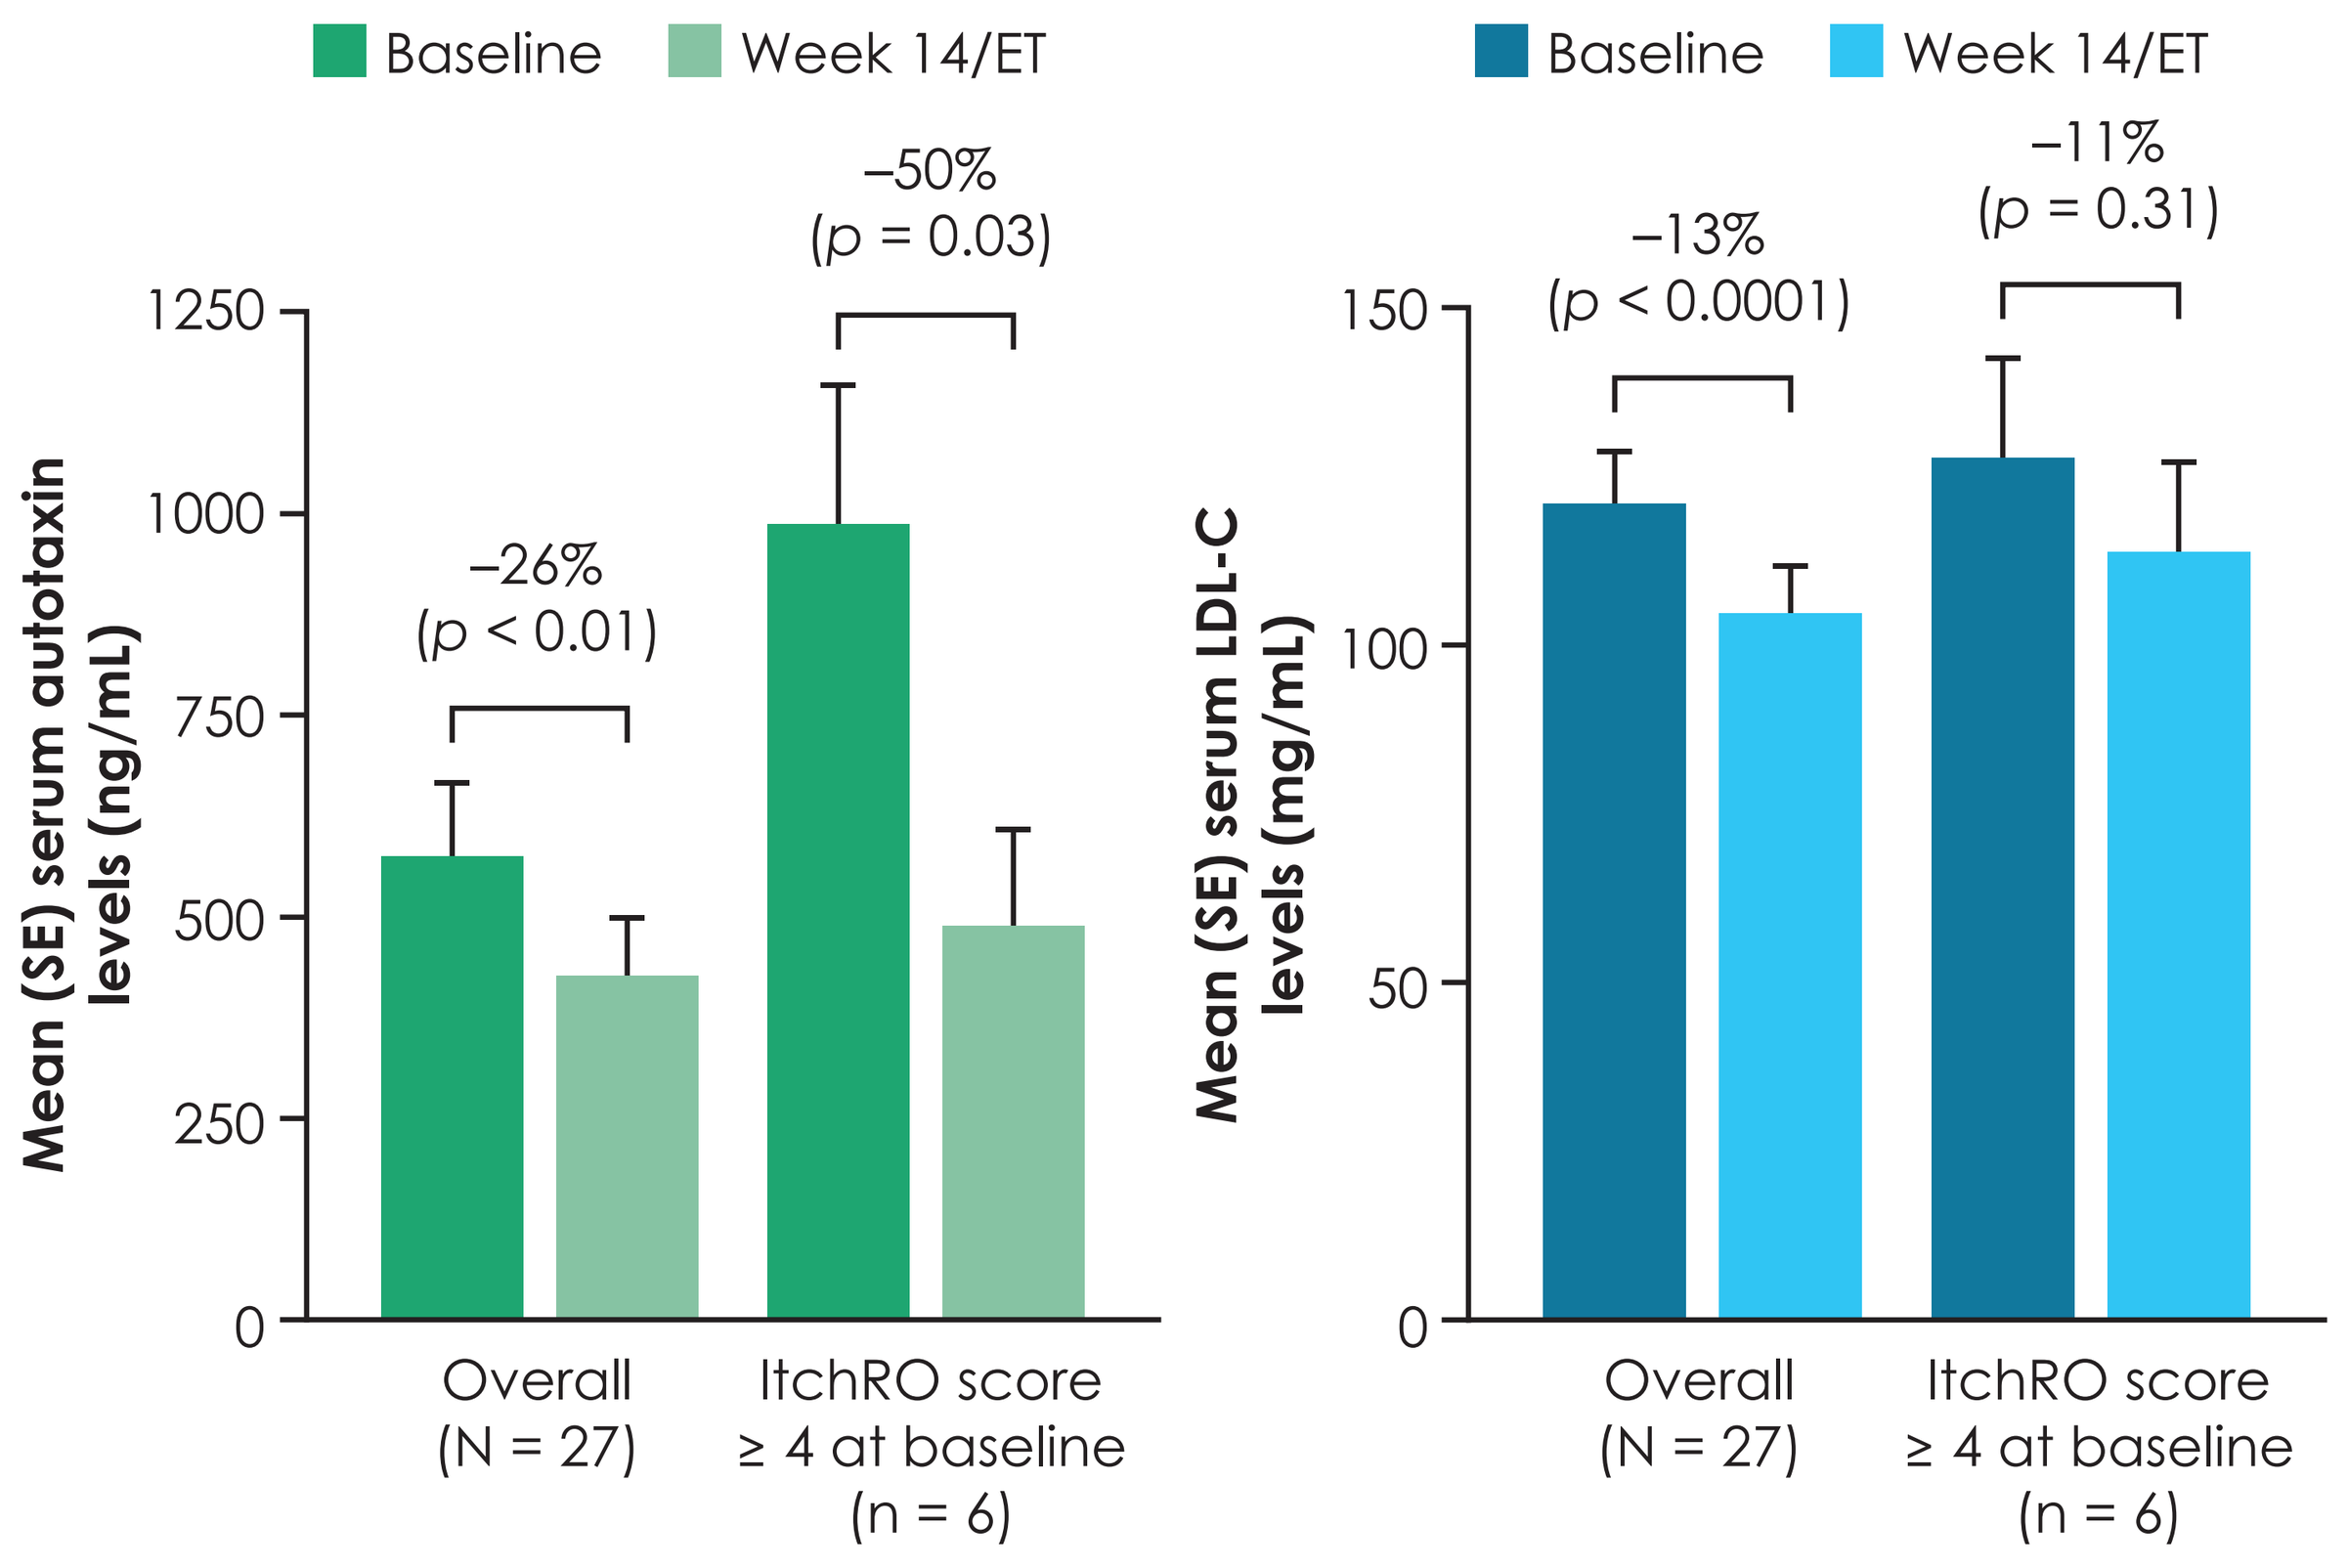
Baseline
Week 14/ET
Mean (SE) serum autotaxin levels (ng/mL)
1250
1000
750
500
250
0
–26%
(p < 0.01)
–50%
(p = 0.03)
Overall
(N = 27)
ItchRO score
≥ 4 at baseline
(n = 6)
Baseline
Week 14/ET
Mean (SE) serum LDL-C levels (mg/mL)
150
100
50
0
–13%
(p < 0.0001)
–11%
(p = 0.31)
Overall
(N = 27)
ItchRO score
≥ 4 at baseline
(n = 6)

Early termination was the last post-baseline value before or within 7 days of the last dose.
ET, early termination; ItchRO, Adult Itch Reported Outcome (0–10 scale); LDL-C, low-density lipoprotein cholesterol; SE, standard error.

Table 2. Change from baseline to week 14/ET in additional laboratory parameters

	Overall (N = 27)		ItchRO daily score ≥ 4 at baseline (n = 6)	
	Mean change (SE)	*p*	Mean change (SE)	*p*
Total cholesterol, mg/dL	−21.2 (4.90)	< 0.001	−32.0 (13.38)	0.06
Conjugated bilirubin, mg/dL	0.19 (0.09)	< 0.05	0.58 (0.280)	0.09
Total bilirubin, mg/dL	0.24 (0.128)	0.07	0.82 (0.332)	0.06
ALT, U/L	10.5 (12.14)	0.10	29.3 (9.78)	0.06
AST, U/L	11.7 (6.57)	0.09	19.0 (15.28)	0.27
ALP, U/L	36.7 (32.88)	0.42	186.7 (108.83)	0.09

Early termination was the last post-baseline value before or within 7 days of the last dose.
ALP, alkaline phosphatase; ALT, alanine aminotransferase; AST, aspartate aminotransferase; ET, early termination; ItchRO, Adult Itch Reported Outcome (0–10 scale); SE, standard error.

Safety Outcomes

- TEAEs occurred in 92.6% of participants: mostly gastrointestinal and generally mild or moderate (**Tables 3** and **4**).
 - Safety outcomes were consistent between the overall population and participants with an ItchRO daily score ≥ 4 at baseline.
- A total of five serious TEAEs were reported in four participants (14.8%).
 - Four events were unrelated to study drug (upper gastrointestinal hemorrhage, melena, joint dislocation, appendicitis) and one event (cholangitis, occurring at day 89) was deemed possibly related.

Table 3. Summary of treatment-emergent adverse events

Participants with at least one TEAE, n (%)	Overall population (N = 27)
Any TEAE	25 (92.6)
Mild intensity	11 (40.7)
Moderate intensity	6 (22.2)
Severe intensity	8 (29.6)
TEAE potentially related to study drug[a]	19 (70.4)
Serious TEAE	4 (14.8)
TEAE leading to study drug discontinuation[b]	2 (7.4)

[a]Any intensity grade; [b]Abdominal discomfort (moderate, related to study drug) and fatigue (severe, possibly related to study drug); occurring in one participant (3.7%) each.
TEAE, treatment-emergent adverse event.

Table 4. Treatment-emergent adverse events of special interest

TEAEs of special interest reported in ≥ 10% of participants, n (%)	Overall population (N = 27)	
	Potentially related to study drug	Total[a]
Gastrointestinal disorders	19 (70.4)	22 (81.5)
Diarrhea	12 (44.4)	14 (51.9)
Nausea	7 (25.9)	9 (33.3)
Abdominal pain	7 (25.9)	8 (29.6)
Abdominal distension	4 (14.8)	4 (14.8)
Abdominal discomfort	2 (7.4)	3 (11.1)
Hepatobiliary disorders	1 (3.7)[b]	4 (14.8)[c]

[a]Includes related and unrelated TEAEs; [b]Cholangitis; [c]Cholangitis (n = 2, 7.4%; one mild and one severe), hepatomegaly (n = 2, 7.4%; both mild), jaundice (n = 1, 3.7%; moderate).
TEAE, treatment-emergent adverse event.

Conclusions

- **In this proof-of-concept trial including 27 adults with PSC who received maralixibat for 14 weeks:**
 - **Statistically significant reductions from baseline in pruritus and levels of sBA and autotaxin were observed.**
 - **These improvements appeared to be greater in participants with an ItchRO daily score ≥ 4 out of 10 at baseline than in the overall population.**
 - **Changes in LDL-C and 7αC4 were consistent with ASBT inhibition.**
 - **TEAEs were generally mild or moderate and were mostly gastrointestinal.**
- **These findings warrant further investigation of ASBT inhibitors for the treatment of adults with PSC.**

Disclosures

George Apostol and Joan Gu are employees of Takeda Pharmaceutical Company Ltd. Alejandro Dorenbaum and Ciara Kennedy are stock shareholders in Mirum Pharmaceuticals. Paul J. Pockros holds a management position at SC Liver Research Consortium, LLC. Pamela Vig holds a management position at Mirum Pharmaceuticals. Bertus Eksteen, Angela Cheung and Lisa M. Forman have no conflicts of interest to declare. The following authors have received compensation for serving as consultants or speakers, or they or the institutions they work for have received research support or royalties from the companies or organizations indicated: Christopher L. Bowlus (BiomX, BMS, ChemomAb, CymaBay Therapeutics, Eli Lilly, Gilead, GSK, Intercept, Novartis, Pliant, Takeda), Cynthia Levy (Alnylam, Cara Therapeutics, CymaBay Therapeutics, Durect, Enanta, Flashlight Therapeutics, Genfit, Genkyotex, Gilead, GSK, High Tide, Intercept, Lilly, Mitsubishi, Novartis, Pliant, Shire Pharmaceuticals [now a member of the Takeda group of companies], Target PharmaSolutions, Zydus), Cynthia A. Moylan (BMS, Genfit, Gilead, Intercept, Madrigal, NGM, Novartis, Novo Nordisk, TaiwanJ), Gideon M. Hirschfield (CymaBay Therapeutics, GSK, Intercept Pharma), Paul J. Pockros (AbbVie, Assembly Biosciences, Conatus Pharmaceuticals, Genfit, Gilead, Intercept, Prometheus), Douglas Thorburn (ChemomAb, CymaBay Therapeutics, Falk, Intercept).

Poster presented at: American Association for the Study of Liver Diseases, The Liver Meeting; Boston, MA, USA; November 8–12, 2019.

References

1. Keller B *et al.* Poster presented at the Falk Symposium 194, October 8–9, 2014, Freiburg, Germany. Poster number 55. 2. Gonzales E *et al. J Hepatol* 2019;70:e119–20. 3. Thompson R *et al. J Hepatol* 2019;70:e131–2. 4. Kremer AE *et al. Hepatology* 2012;56:1391–400.

Acknowledgments

Maralixibat is owned by Mirum Pharmaceuticals Inc. This study was funded by the previous owner, Shire Pharmaceuticals Inc, now a member of the Takeda group of companies. Under the direction of the authors, Karis Vaughan PhD, an employee of PharmaGenesis Cardiff, Cardiff, UK, provided writing assistance for this publication. Formatting, proofreading, copy editing and fact checking assistance was also provided by PharmaGenesis Cardiff. Writing and editorial assistance was funded by Mirum Pharmaceuticals in accordance with Good Publication Practice 3 (GPP3) guidelines.