Psychometric evaluation of the Adult Itch Reported Outcome tool, a worst itch numeric rating scale in adults with cholestatic liver disease

Brandon Foster,[1] David Andrae,[1] Pamela Vig,[2] Andrew McKibben,[2] Christopher L Bowlus[3]

[1]Endpoint Outcomes, Boston, MA, USA; [2]Mirum Pharmaceuticals, Foster City, CA, USA; [3]University of California Davis School of Medicine, Sacramento, CA, USA

FRI131

Introduction

- Primary biliary cholangitis (PBC) and primary sclerosing cholangitis (PSC) are chronic, progressive cholestatic liver diseases that cause deterioration of bile ducts and often lead to liver transplantation.[1,2]
- The most common symptoms of PBC and PSC include fatigue and pruritus, which may significantly impair quality of life.[1–3]
- The severity of pruritus can be measured with the Adult Itch Reported Outcome (ItchRO) tool, an 11-point, patient-completed, single-item, worst itch numeric rating scale (NRS).
- Maralixibat (MRX), a selective inhibitor of the apical sodium-dependent bile acid transporter, is in clinical development for the treatment of cholestatic liver disease.[4–7]
- The Adult ItchRO tool was originally developed using qualitative methods and cognitive debriefing in patients with PBC, and was a secondary endpoint in two studies of MRX in PBC (CLARITY; NCT01904058) and PSC (CAMEO; NCT02061540).[4,8]

Aim

- To assess and validate the scoring and psychometric properties of the Adult ItchRO tool for pruritus in adult patients with cholestatic liver disease in a *post hoc* analysis of data from the CLARITY and CAMEO studies.

Methods

Study population

- CLARITY was a 13-week, randomized, double-blind, placebo-controlled study with an intention-to-treat sample of 66 patients with PBC.
- CAMEO was a 14-week, pilot, open-label study in 27 patients with PSC.

Adult ItchRO tool

- The Adult ItchRO tool was completed twice daily (morning [AM] and evening [PM]) using electronic (e-) diaries. Patients were asked to rate their worst itch from the time they went to bed until waking up (AM diary), and from waking up until the time of completion (PM diary), using an 11-point NRS (0 = 'no itch' to 10 = 'worst possible itch').
- In the CLARITY study, the e-diaries were completed at screening, baseline, and Weeks 1, 2, 3, 4, 8, and 13; and in the CAMEO study, at screening, baseline, and Weeks 1, 2, 3, 4, 5, 6, 10, and 14.
- At each time point, the Adult ItchRO e-diary was completed daily for 7 days and the average of the daily worst rating during this period was calculated (7-day score).

Other numerical rating scales used to validate the Adult ItchRO tool

- Covalidators for the Adult ItchRO tool included 5D-Itch Scale, PBC-40, Medical Outcomes Study (MOS)-Sleep Scale, and Patient Impression of Change in Itchiness (PIC-Itch).[9–11]

Psychometric validation

- Due to different study designs, analyses of the CLARITY and CAMEO studies were conducted separately.

Exchangeability of the AM and PM items

- The exchangeability of patients' AM and PM e-diaries during baseline week was investigated using a longitudinal mixed model.

Assessment of score quality

- Concurrent validity for the Adult ItchRO scores was examined by calculating correlations between the Adult ItchRO and the 5D-Itch Scale total scores, the six domains of the PBC-40, and the six dimensions of the MOS-Sleep Scale.
- The test-retest reliability (TRR) of the Adult ItchRO scores was investigated by calculating the two-way random intraclass correlation coefficients (*ICCs* [2,1])[12] using the scores from the week prior to baseline.

Analysis of minimal clinically important change in the Adult ItchRO score

- Anchor-based estimates were generated for the PBC study by calculating summaries of the change in Adult ItchRO scores from baseline to Week 13 within strata of patients' PIC-Itch ratings.
- Empirical cumulative density function (CDF) curves were generated for the Adult ItchRO scores and were stratified by PIC-Itch ratings.

Results

Adult ItchRO scores

- Figures 1 and 2 show individual patient profiles for AM and PM Adult ItchRO scores.
- At baseline, patients with PBC had more severe ratings for their pruritus (CLARITY; Figure 1) than patients with PSC (CAMEO; Figure 2).
- In patients with PBC with pruritus, the most utilized rating categories were 'severe' to 'most severe'.
- Scores improved after MRX initiation in patients with PBC with less-severe to moderately severe pruritus at baseline (Figure 1).
- Scores improved after MRX initiation in patients with PSC with moderate and severe pruritus at baseline (Figure 2).

Figure 1. CLARITY[a] longitudinal responses to the Adult ItchRO from the screening period to Week 13

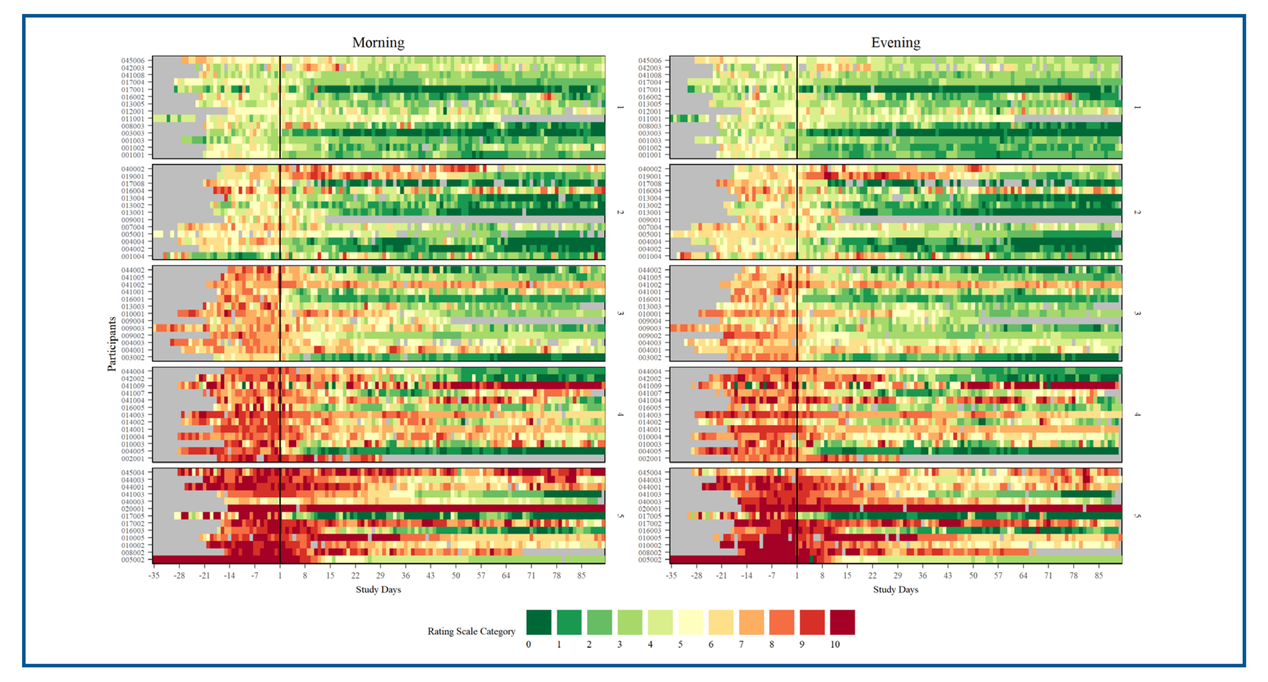
Morning
Evening
Participants
Study Days
Rating Scale Category

[a]Patients with PBC
ItchRO, Itch Reported Outcome; PBC, primary biliary cholangitis
Plots are faceted by quantiles for the weekly average of the daily worst (i.e. highest rated) Adult ItchRO score and arranged in ascending order.
The vertical line in the plots indicates baseline and the introduction of treatment.
Changes in the intensity of pruritus across time can be tracked by monitoring changes in the color of each patient's response vector (red = worse; green = better)

Figure 2. CAMEO[a] longitudinal responses to the Adult ItchRO from the screening period to Week 14

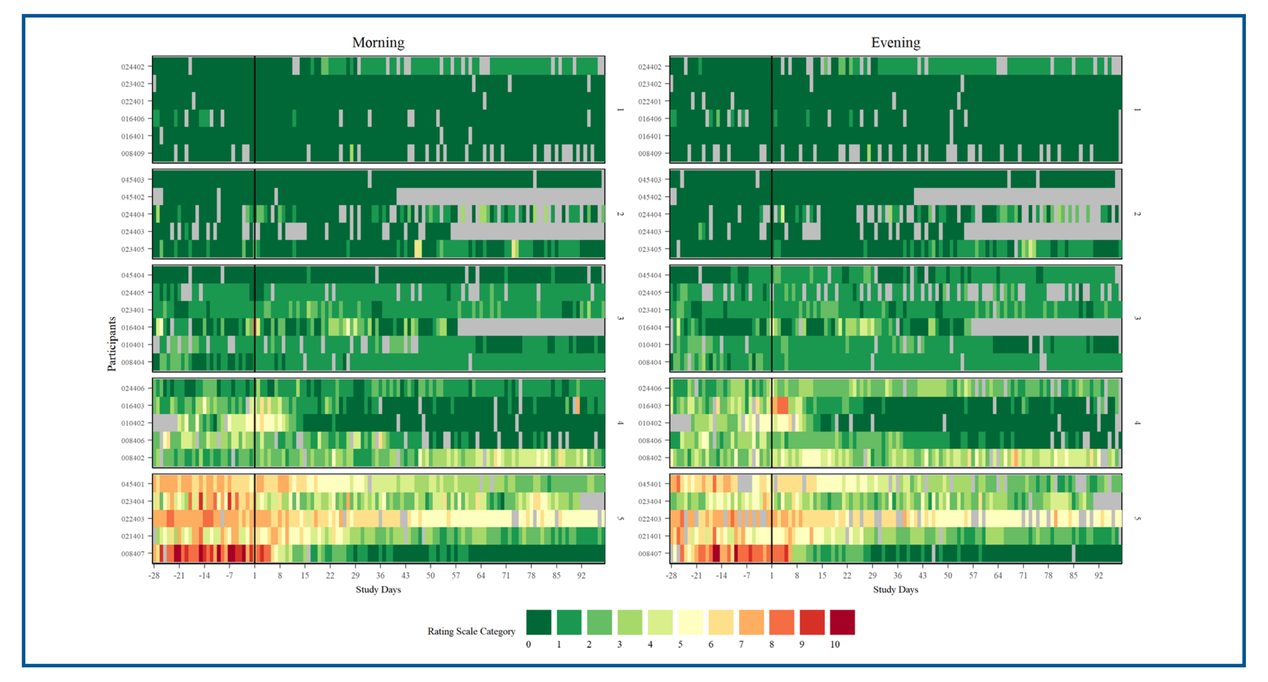
Morning
Evening
Participants
Study Days
Rating Scale Category

[a]Patients with PSC
ItchRO, Itch Reported Outcome; PSC, primary sclerosing cholangitis
Plots are faceted by quantiles for the weekly average of the daily worst (i.e. highest rated) Adult ItchRO score and arranged in ascending order.
The vertical line in the plots indicates baseline and the introduction of treatment.
Changes in the intensity of pruritus across time can be tracked by monitoring changes in the color of each patient's response vector (red = worse; green = better)

Exchangeability of the AM and PM items

- Linear mixed models showed that patients' AM and PM ratings of the Adult ItchRO tool (Figures 1 and 2) were exchangeable (i.e. there were no differences); thus, a weekly average score of the daily worst itch rating was calculated and used in subsequent reliability and validity analyses.

Concurrent validity

- The Pearson correlation coefficients for the Adult ItchRO and the 5D-Itch Scale scores, PBC-40 domains, and MOS-Sleep dimensions in patients with PBC and PSC are shown in Figures 3 and 4, respectively.
- The strongest association in both studies was with the 5D-Itch Scale total score ($r = 0.59$ [patients with PBC] and $r = 0.95$ [patients with PSC]).
- Correlations were found between the Adult ItchRO scores and other covalidators, including the PBC-Itch ($r = 0.51$ and 0.85, respectively), PBC-Emotion ($r = 0.42$ and 0.57, respectively), and MOS-Disturbance ($r = 0.40$ and 0.60, respectively).

Figure 3. CLARITY[a] heatmap of the Pearson correlation coefficients for convergent and divergent validity of the Adult ItchRO scores

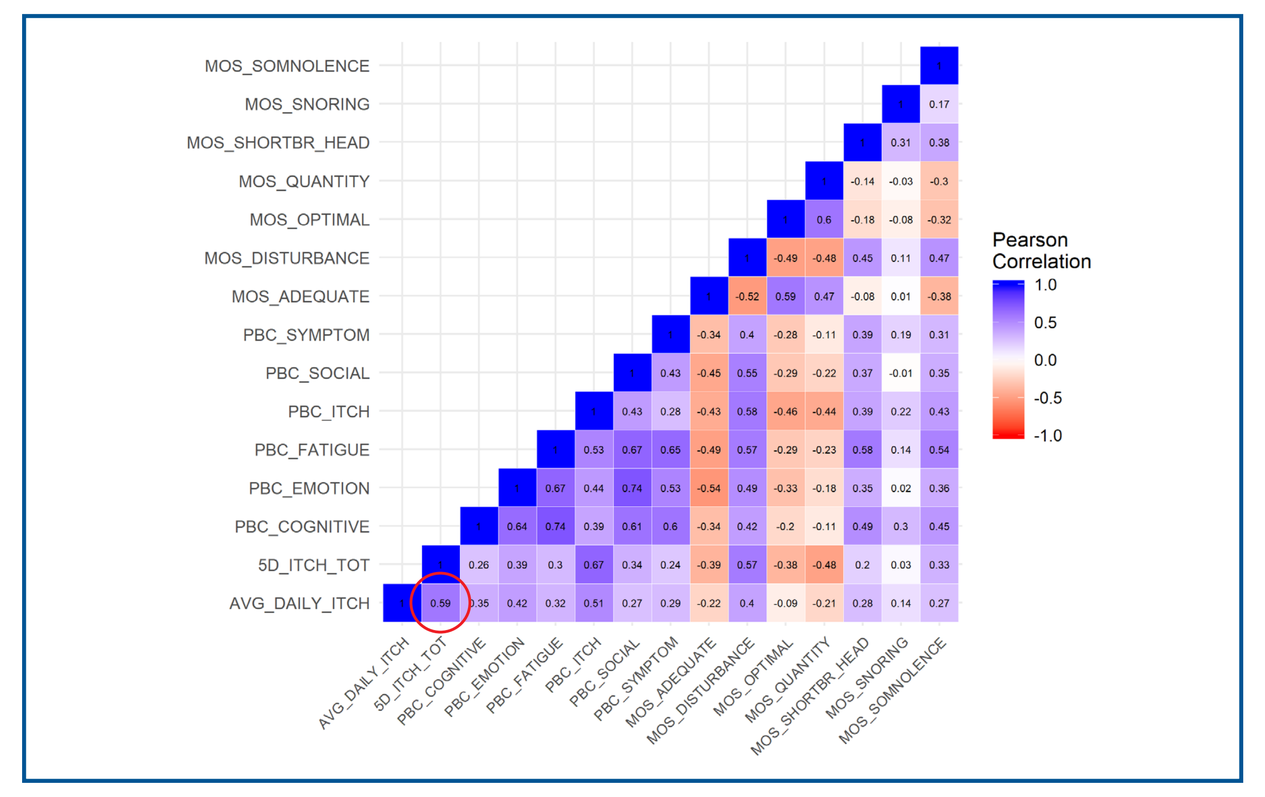
Pearson Correlation

[a]Patients with PBC
ItchRO, Itch Reported Outcome; PBC, primary biliary cholangitis
The average daily Adult ItchRO scores were correlated in the expected direction with several covalidating measures

Figure 4. CAMEO[a] heatmap of the Pearson correlation coefficients for convergent and divergent validity of the Adult ItchRO scores

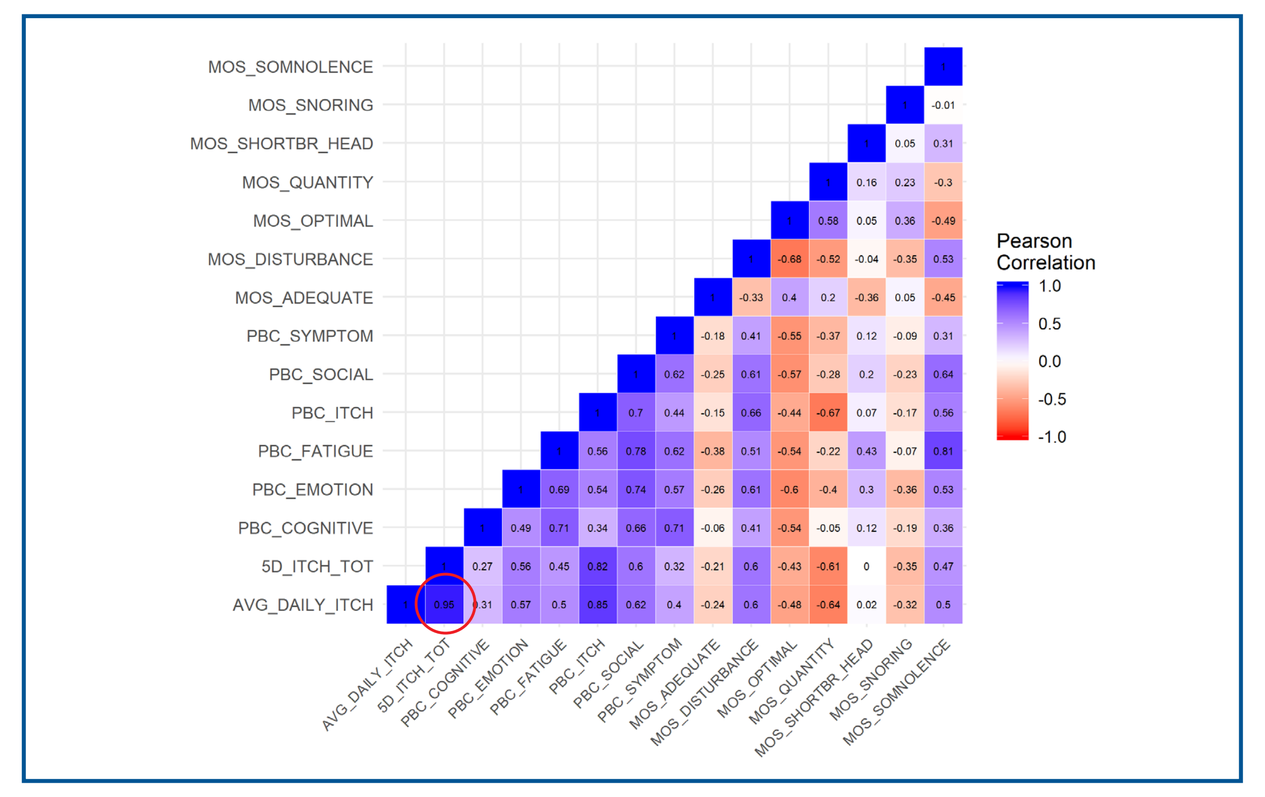
Pearson Correlation

[a]Patients with PSC
ItchRO, Itch Reported Outcome; PSC, primary sclerosing cholangitis
The average daily Adult ItchRO scores were correlated in the expected direction with several covalidating measures

Test-retest reliability

- A two-way random *ICC* (2,1) ≥ 0.70 was considered sufficient evidence for demonstrating TRR; estimates for TRR were 0.93 (95% confidence interval [CI] 0.89, 0.96) for patients with PBC and 0.98 (95% CI 0.95, 0.99) for patients with PSC.

Minimal clinically important change

- The estimated standard error of measurement (SEM) in both studies was < 0.50 (Table 1). A threshold of 2 SEMs was used to define minimal detectable change.
 - Results suggest that change from baseline in the Adult ItchRO score of as little as –1.0 would likely exceed the measurement error of the Adult ItchRO and be indicative of real underlying change in pruritus severity.
- Most patients with PBC showed elevated ItchRO scores at baseline and, overall, most change from baseline scores exceeded criteria for a minimal detectable difference. Not all patients with PSC had elevated scores at baseline, but those who did responded similarly to patients with PBC. However, a smaller percentage of patients with PSC showed change from baseline that exceeded the criteria for minimal detectable difference, which was likely due to the ItchRO scores at baseline.

Table 1. Number of patients in the CLARITY[a] and CAMEO[b] studies with change scores ≤ 2 SEMs at study endpoint

Study	SEM	Below 2 SEMs	Not below 2 SEMs	N
CLARITY	0.491	57	9	66
CAMEO	0.408	8	19	27

[a]Patients with PBC; [b]patients with PSC
PBC, primary biliary cholangitis; PSC, primary sclerosing cholangitis; SEM, standard error of measurement
The criteria of ≤ 2 SEMs (i.e. lower interval of the SEM) accommodates the research design where patients were expected to proceed from worst itch at baseline (i.e. response to the higher rating categories of the Adult ItchRO) to lower responses at Week 13 (CLARITY) or Week 14 (CAMEO)

- Approximately 50% of patients with PBC (37/66) recorded that PIC-Itch was 'much better' at Week 13 with an average decrease in the Adult ItchRO score of –5.09 (Table 2).
- Average Adult ItchRO change scores within the two PIC-Itch categories used to define the anchors for establishing meaningful change were 'no change' (M = –0.89) and 'a little better' (M = –2.63).
- The estimate for meaningful change in the Adult ItchRO score was –1.74 (i.e. the difference between –0.89 and –2.63).

Table 2. Descriptive statistics for Adult ItchRO change scores at Week 13 (endpoint) of CLARITY[a] stratified by the PIC-Itch

PIC-Itch	N	Mean	SD
Much better	37	–5.09	2.05
Better	4	–3.36	0.89
A little better	10	–2.63	1.41
No change	10	–0.89	1.13
Worse	2	–0.21	0.30
Much worse	1	0.71	NA
Missing	2	–1.71	1.82

[a]Patients with PBC
ItchRO, Itch Reported Outcome; NA, not applicable; PBC, primary biliary cholangitis; PIC-Itch, Patient Impression of Change in Itchiness; SD, standard deviation

- CDF plots for change from baseline scores in the Adult ItchRO stratified by PIC-Itch ratings (Figure 5) showed CDF curves were monotonically separated in the expected order (i.e. 'much better', 'better', 'a little better', and 'no change'), indicating patients' level of change in the Adult ItchRO scores within each of these groups tended to be distinct from the other groups defined by the PIC-Itch.

Figure 5. CLARITY[a] empirical cumulative density curves of change from baseline to Week 13 in the Adult ItchRO scores (endpoint) stratified by the PIC-Itch

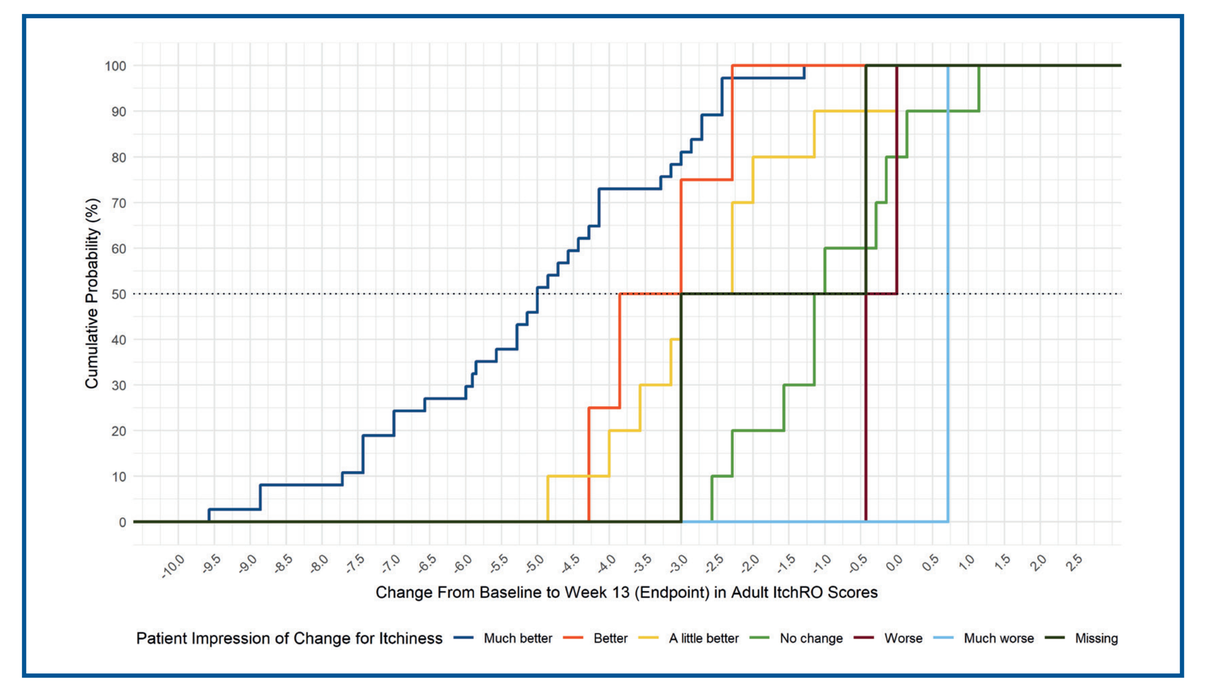
Cumulative Probability (%)
Change From Baseline to Week 13 (Endpoint) in Adult ItchRO Scores
Patient Impression of Change for Itchiness
Much better
Better
A little better
No change
Worse
Much worse
Missing

[a]Patients with PBC
ItchRO, Itch Reported Outcome; PBC, primary biliary cholangitis; PIC-Itch, Patient Impression of Change in Itchiness

Conclusions

- The Adult ItchRO is an 11-point scale, measured AM and PM, that has good psychometric properties.
- Scores from the measure showed satisfactory convergent validity with covalidators, ability to distinguish between known groups, and strong TRR.
- Distribution-based approaches showed the measure could detect changes as small as 1 point, while the anchor-based estimate for meaningful change was –1.74.
- The Adult ItchRO was shown to be strongly correlated with the 5D-Itch Scale and demonstrated favorable correlations with other quality of life measures of emotion and sleep disturbance.
- The Adult ItchRO may be a suitable endpoint for clinical studies investigating potential treatments for patients with PBC and PSC.

Contact information
Brandon Foster, brandon.foster@endpointoutcomes.com

Poster presented at The Digital International Liver Congress™ 2020, 27–29 August 2020

References

1. Lee Y-M, Kaplan MM. Primary sclerosing cholangitis. *N Engl J Med* 1995;332(14):924–933; 2. Chapman RW, Arborgh BA, Rhodes JM, *et al.* Primary sclerosing cholangitis: a review of its clinical features, cholangiography, and hepatic histology. *Gut* 1980;21(10):870–877; 3. Kaplan MM. Primary Biliary Cirrhosis. In: Lennarz WJ, Lane DM (Editors-in-Chief). *Encyclopedia of Biological Chemistry.* 2nd edn. Cambridge, MA: Academic Press; 2013:561–565; 4. Mayo MJ, Pockros PJ, Jones D, *et al.* A randomized, controlled, phase 2 study of maralixibat in the treatment of itching associated with primary biliary cholangitis. *Hepatol Commun* 2019;3(3):365–381; 5. Baker A, Kelly D, McClean P, *et al.* A phase 2, randomized, placebo-controlled study (IMAGO) of LUM001, a novel inhibitor of the apical sodium-dependent bile acid transporter (ASBT), in paediatric patients with alagille syndrome (ALGS). *J Hepatol* 2015;62(S2):S259; 6. Gonzales EM, Sturm E, Stormon M, *et al.* Durability of treatment effect with long-term maralixibat in children with alagille syndrome: 4-year safety and efficacy results from the ICONIC study. Presented at AASLD, The Liver Meeting®, 8–12 November 2019, Boston, MA, USA; 7. Thompson RJ, Jaecklin T, Peetz C; LUM001-501 INDIGO Study Group. Genotype and dose-dependent response to maralixibat in patients with bile salt export pump deficiency. Presented at AASLD, The Liver Meeting®, 8–12 November 2019, Boston, MA, USA; 8. Bowlus CL, Eksteen B, Cheung A, *et al.* Safety and efficacy of maralixibat in patients with primary sclerosing cholangitis: an open-label proof-of-concept study. Presented at AASLD, The Liver Meeting®, 8–12 November 2019, Boston, MA, USA. Poster presentation; 9. Elman S, Hynan LS, Gabriel V, *et al.* The 5-D itch scale: a new measure of pruritus. *Br J Dermatol* 2010;162(3):587–593; 10. Jacoby A, Rannard A, Buck D, *et al.* Development, validation, and evaluation of the PBC-40, a disease specific health related quality of life measure for primary biliary cirrhosis. *Gut* 2005;54(11):1622–1629; 11. Hays RD, Martin SA, Sesti AM, *et al.* Psychometric properties of the Medical Outcomes Study Sleep measure. *Sleep Med* 2005;6(1):41–44; 12. Shrout PE, Fleiss JL. Intraclass correlations: uses in assessing rater reliability. *Psychol Bull* 1979;86(2):420–428

Acknowledgments
Maralixibat is owned by Mirum Pharmaceuticals Inc.
The psychometric work was conducted by Endpoint Outcomes and funded by Mirum Pharmaceuticals Inc.

Editorial assistance was provided by Alpharmaxim Healthcare Communications and funded by Mirum Pharmaceuticals Inc.