Clinical Benefits With Maralixibat for Patients With Alagille Syndrome Are Durable Through 7 Years of Treatment: Data From the MERGE Study

Poster THU-094

Binita M. Kamath,[1] Emmanuel Gonzalès,[2] Deirdre Kelly,[3] Douglas B. Mogul,[4] Will Garner,[4] Pamela Vig,[4] Emmanuel Jacquemin[2]

[1]The Hospital for Sick Children and the University of Toronto, Division of Gastroenterology, Hepatology and Nutrition, Toronto, Ontario, Canada; [2]Service d'Hépatologie et de Transplantation Hépatique Pédiatriques, Centre de Référence de l'Atrésie des Voies Biliaires et des Cholestases Génétiques (AVB-CG), FSMR FILFOIE, ERN RARE LIVER, Hôpital Bicêtre, AP-HP, Faculté de Médecine Paris-Saclay, Le Kremlin-Bicêtre, and Inserm U1193, Hépatinov, Université Paris-Saclay, Orsay, France; [3]Liver Unit, Birmingham Women's & Children's Hospital NHS Trust and University of Birmingham, Birmingham, United Kingdom; [4]Mirum Pharmaceuticals, Inc., Foster City, California, USA

Introduction

- Alagille syndrome (ALGS) is a rare, debilitating, autosomal dominant disorder that presents with a broad range of clinical manifestations.[1]
 - The key clinical manifestations include cholestasis, pruritus, failure to thrive, xanthomas, and progressive liver disease, all of which can lead to liver transplant or death.
- Maralixibat is a minimally absorbed ileal bile acid transporter inhibitor that prevents enterohepatic bile acid recirculation and is approved for the treatment of cholestatic pruritus in patients with ALGS ≥2 months of age in the EU.[2]
- Improvements in pruritus, sBA, and height were observed in prior clinical trials of maralixibat in participants with ALGS.[3,4]
 - In ICONIC, responses were durable for up to ~4 years.[3]
 - In IMAGO and its extension study IMAGINE and in ITCH and its extension study IMAGINE-II, responses were durable for up to ~1.5 years.[4]
- Participants from ICONIC, IMAGINE, and IMAGINE-II were invited to enrol in the MERGE study for additional follow-up to assess the long-term durability of response to maralixibat.[5]

Objective

- To report on the efficacy of maralixibat in participants with ALGS with additional long-term follow-up from MERGE, including some participants who received treatment for 7 years.

Methods

- All participants from ICONIC, IMAGINE and IMAGINE-II were included in the analysis; details of the study designs have been presented previously.
- Impact of maralixibat was assessed for pruritus, sBA, height and weight Z-scores, ALT, AST, and total and direct bilirubin.[a]
 - Pruritus was measured using Itch-Reported Outcome (Observer) (ItchRO[Obs]), which is a 0 to 4 scale where 0 = no itch, 1 = mild, 2 = moderate, 3 = severe, and 4 = very severe.[6] A ≥1-point reduction in ItchRO(Obs) is considered clinically meaningful.
- Change from Baseline (CFB) was assessed by comparing median (Q1, Q3) values from enrolment in the initial trial (ie, ICONIC, IMAGO, or ITCH) with data from the visit in MERGE that best aligned with an annual visit.

[a]ALT and bilirubin were measured in serum.

Results

Table 1. Key Demographics and Baseline Characteristics

Variable[a]	Integrated population (N=86)
Age, y	5 (2, 9)
Sex, male, %	57.0
Pruritus, ItchRO(Obs)	2.7 (2.1, 3.1)
Total sBA, μmol/L	194 (83, 363)
UDCA usage, n (%)	68 (79.1)
ALT, U/L	140 (95, 196)
AST, U/L	133 (89, 187)
Total bilirubin, μmol/L	50.5 (17.1, 138.5)
Direct bilirubin, μmol/L	43.6 (12.0, 128.3)
Height Z-score[b]	-1.6 (-2.3, -0.9)
Weight Z-score[b]	-1.4 (-2.2, -0.7)

[a]All data are median (Q1, Q3) unless otherwise indicated. [b]Height and weight Z-scores are based on a participant's sex and age at the Baseline visit. The World Health Organization growth charts were used to derive Z-scores for participants younger than 24 months, and the Centers for Disease Control and Prevention growth charts were used to derive Z-scores for participants aged 24 months or older.

Improvements in sBA Persist for up to 7 Years of Maralixibat Treatment

Table 2. CFB in Clinical Parameters of Interest Over Time[a]

Parameter	Year 1 (n=76)	Year 2 (n=49)	Year 3 (n=45)	Year 4 (n=42)	Year 5 (n=40)	Year 6 (n=34)	Year 7 (n=23)
ItchRO(Obs)	-1.6 (-2.1, -0.8)	-1.0 (-1.9, -0.3)	-1.0 (-2.0, -0.3)	-1.1 (-2.0, -0.5)	-2.0 (-2.9, -1.3)	-1.3 (-1.9, -0.6)	-2.1 (-2.6, -1.6)
	<0.001	**<0.001**	**<0.001**	**<0.001**	**<0.001**	**<0.001**	**0.004**
sBA, μmol/L	-53 (-146, 8)	-62 (-137, 2)	-60 (-134, 0)	-61 (-152, -34)	-77 (-182, -21)	-113 (-235, -24)	-121 (-231, -41)
	<0.001	**<0.001**	**<0.001**	**<0.001**	**<0.001**	**<0.001**	**<0.001**
ALT, U/L	23 (-3, 64)	42 (-21, 100)	33 (-2, 74)	36 (-4, 98)	54 (8, 116)	43 (14, 131)	72 (5, 87)
	<0.001	**0.002**	**<0.001**	**0.006**	**<0.001**	**<0.001**	**0.009**
AST, U/L	14 (-7, 35.5)	21 (-12, 58)	19 (-9, 46)	13 (-23, 59)	31 (-14, 96)	3 (-11, 63)	26 (-10, 70)
	0.001	**0.01**	**0.02**	**0.04**	**0.001**	0.09	**0.04**
Total bilirubin, μmol/L	0.0 (-9.4, 8.6)	-1.7 (-13.7, 6.8)	0.0 (-18.8, 6.8)	-2.6 (-29.1, 3.4)	-3.4 (-18.0, 4.1)	-1.7 (-13.7, 6.7)	1.7 (-11.4, 9.2)
	0.89	0.14	0.19	**0.02**	0.10	0.33	0.71
Direct bilirubin, μmol/L	0.0 (-6.0, 1.7)	-1.7 (-10.3, 1.7)	-3.4 (-12.0, 1.7)	-5.1 (-18.8, 0.0)	-1.7 (-13.7, 1.7)	-3.4 (-10.3, 1.7)	-1.3 (-12.0, 2.7)
	0.08	**0.008**	**0.008**	**<0.001**	**0.02**	**0.01**	0.56
Height Z-score	0.1 (-0.1, 0.3)	0.2 (0.0, 0.5)	0.4 (0.0, 0.6)	0.3 (0.0, 0.8)	0.4 (-0.1, 0.9)	0.5 (0.0, 1.1)	0.8 (0.0, 1.2)
	<0.001	**<0.001**	**<0.001**	**<0.001**	**0.002**	**<0.001**	**0.002**
Weight Z-score	0.1 (-0.2, 0.4)	0.2 (-0.4, 0.5)	0.1 (-0.2, 0.5)	0.3 (-0.3, 0.5)	0.2 (-0.4, 1.2)	0.4 (-0.4, 1.3)	0.1 (-0.5, 0.9)
	0.26	0.17	0.08	0.15	0.11	0.10	0.51

[a]Data shown are median (Q1, Q3) CFB; *P* value. *P* value is from a signed-rank test that used a within-group comparison to assess whether the CFB is significantly different from 0 (no change).

Conclusions

- Of those participants who remained on maralixibat therapy out to 7 years, nearly all experienced benefit, including reductions in pruritus and sBA.
- Improvements in height were also durable and observed out to 7 years.
- Reductions in direct bilirubin were observed in participants who remained on therapy, as well as slightly increased liver transaminases.

Pruritus Response Persists for up to 7 Years of Maralixibat Treatment

Figure 1. Maralixibat Exposure and ItchRO(Obs) Response by Participant

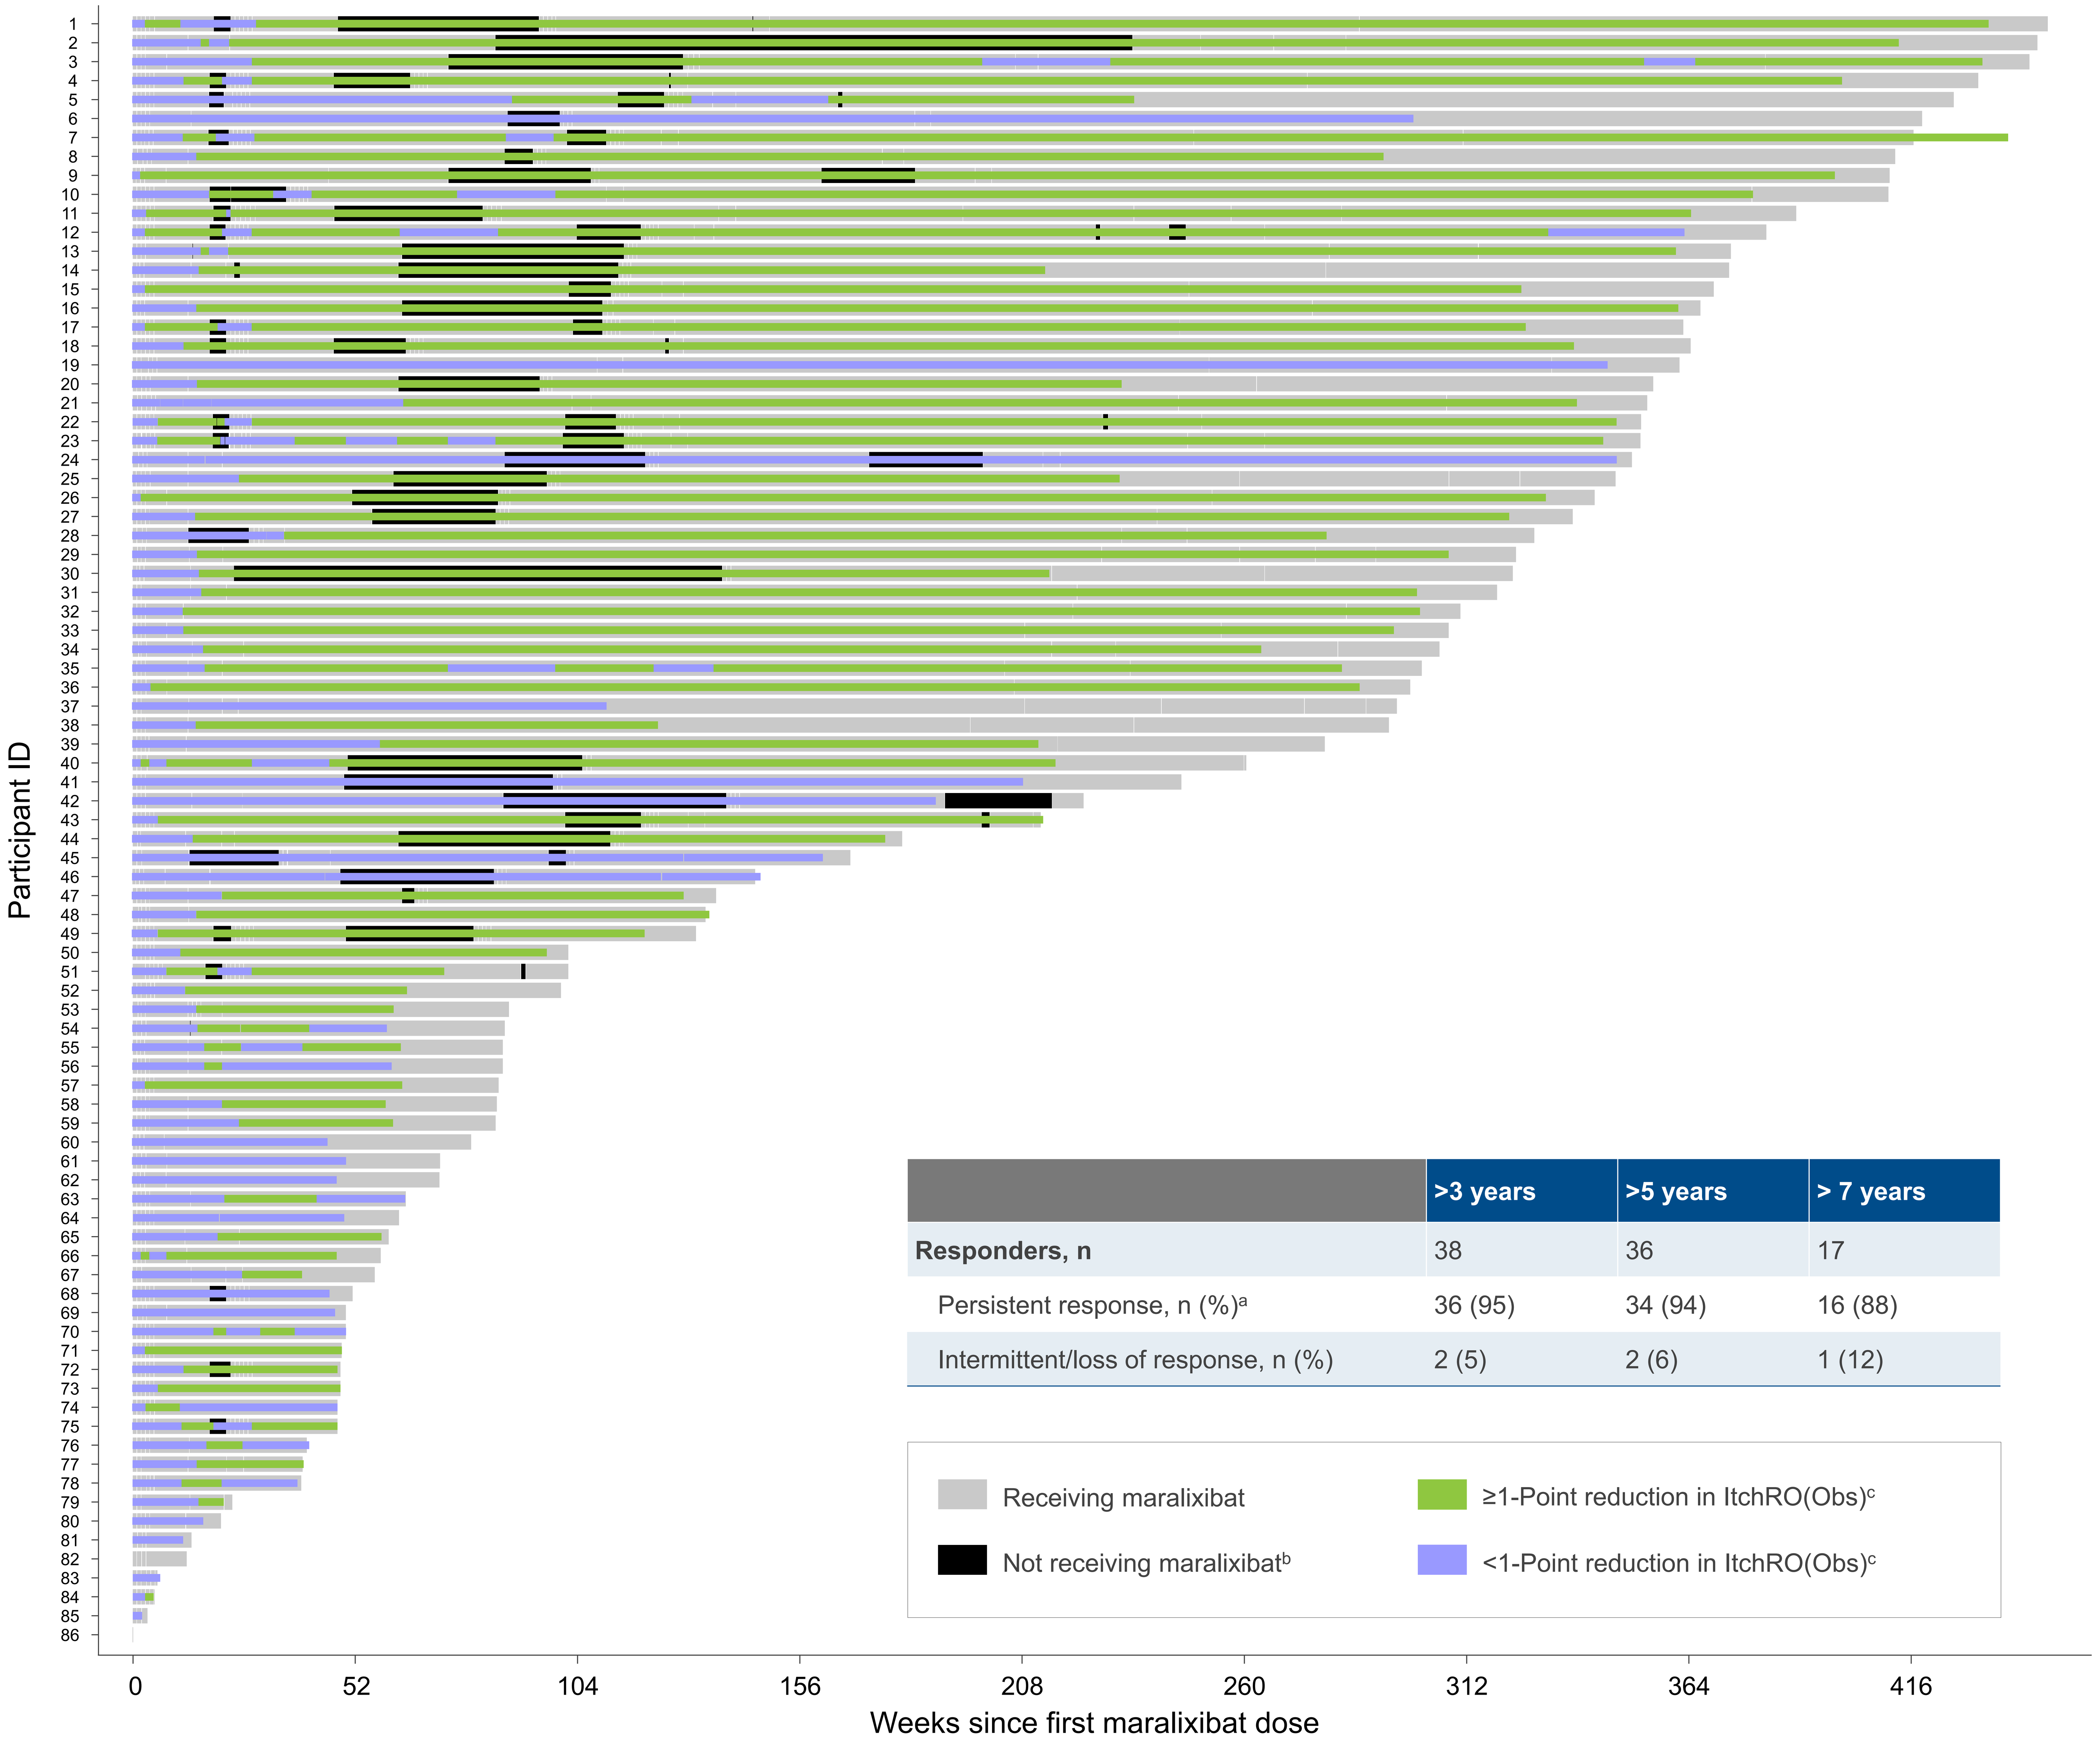
Participant ID
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28 29 30 31 32 33 34 35 36 37 38 39 40 41 42 43 44 45 46 47 48 49 50 51 52 53 54 55 56 57 58 59 60 61 62 63 64 65 66 67 68 69 70 71 72 73 74 75 76 77 78 79 80 81 82 83 84 85 86
0 52 104 156 208 260 312 364 416
Weeks since first maralixibat dose
Receiving maralixibat
Not receiving maralixibat[b]
≥1-Point reduction in ItchRO(Obs)[c]
<1-Point reduction in ItchRO(Obs)[c]

	>3 years	>5 years	> 7 years
Responders, n	38	36	17
Persistent response, n (%)[a]	36 (95)	34 (94)	16 (88)
Intermittent/loss of response, n (%)	2 (5)	2 (6)	1 (12)

[a]Response was evaluated among individuals who had an initial response and was considered durable if >85% of measurements showed a reduction of ≥1 point in ItchRO(Obs). [b]Gaps in maralixibat exposure are due to treatment interruption or participants receiving placebo during Weeks 18 to 22 in ICONIC. [c]Reduction in ItchRO(Obs) relative to baseline (first dose of maralixibat).

Abbreviations

ALGS, Alagille syndrome; ALT, alanine aminotransferase; AST, aspartate aminotransferase; CFB, change from Baseline; ItchRO(Obs), Itch-Reported Outcome (Observer); sBA, serum bile acid; UDCA, ursodeoxycholic acid.

Disclosures

BMK reports grants or contracts from Mirum Pharmaceuticals, Inc. and Albireo and consulting fees from Mirum Pharmaceuticals, Inc., Albireo, and Audentes. EG reports consulting fees from CTRS, Vivet, Mirum Pharmaceuticals, Inc., and Albireo and fees for participation on a data safety monitoring board or advisory board from Mirum Pharmaceuticals, Inc. and Albireo. DK reports grants from Albireo, AbbVie, Gilead Sciences, Mirum Pharmaceuticals, Inc., and Intercept (for clinical trials); consulting fees from Albireo, Alnylam, Mirum Pharmaceuticals, Inc., Intercept, Takeda, Freeline, GSK, Orphalan, and AstraZeneca; and honoraria from Mirum Pharmaceuticals, Inc. and Albireo. DBM, WG, and PV are employees of and shareholders in Mirum Pharmaceuticals, Inc. EJ reports consulting fees from Laboratoire CTRS and Vivet Therapeutics.

Acknowledgments

The authors would like to thank the clinical trial participants, their families, and investigators for their participation in this study. Maralixibat is owned by Mirum Pharmaceuticals, Inc. This analysis was funded by Mirum Pharmaceuticals, Inc. Medical writing and editorial support for the development of this poster was provided by Precision AQ in Yardley, Pennsylvania, USA, which was funded by Mirum Pharmaceuticals, Inc.

References

1. Saleh M, et al. *Appl Clin Genet.* 2016;9:75-82. **2.** LIVMARLI® (maralixibat) [summary of product characteristics]. Amsterdam, Netherlands; Mirum Pharmaceuticals International B.V. Jan 2024. **3.** Gonzales E, et al. *Lancet.* 2021;398:1581-1592. **4.** Shneider BL, et al. *Hepatol Commun.* 2022;6:1922-1933. **5.** ClinicalTrials.gov identifier: NCT04168385. Updated January 30, 2024. Accessed May 1, 2024. https://www.clinicaltrials.gov/study/NCT04168385. **6.** Kamath BM, et al. *Hepatol Commun.* 2020;4:1012-1018.

Presented at European Association for the Study of the Liver (EASL) Congress; 5-8 June 2024; Milan, Italy