Bile Acid Subspecies Are Correlated With Pruritus and Bilirubin Improvement in PFIC Patients Treated With Maralixibat: Data From MARCH and MARCH-ON

Poster 4436

Henkjan J. Verkade,[1] Alexander G. Miethke,[2] Simon P. Horslen,[3] Douglas B. Mogul,[4] Tiago Nunes,[4] Cory Kostrub,[4] Josephine Shelton,[4] Will Garner,[4] Pamela Vig,[4] Richard J. Thompson[5]

[1]Beatrix Children's Hospital University Medical Center, Groningen, The Netherlands; [2]Cincinnati Children's Hospital Medical Center, Cincinnati, Ohio; [3]UPMC Children's Hospital of Pittsburgh, Pittsburgh, Pennsylvania; [4]Mirum Pharmaceuticals, Inc., Foster City, California; [5]King's College London, London, United Kingdom

Introduction

- Progressive familial intrahepatic cholestasis (PFIC) is a group of genetic disorders that result in disrupted bile composition and chronic cholestasis.[1]
 - Key clinical manifestations include debilitating pruritus, impaired growth, reduced quality of life, and progressive liver disease with an eventual need for liver transplantation.[1]
- Pruritus in cholestatic liver diseases, such as PFIC, may result in part from accumulation of toxic bile acids (BAs).[2,3]
 - BAs, including hydrophobic secondary BAs, have markedly higher concentrations in patients with PFIC and are potent inflammatory agents that induce apoptosis in hepatocytes.[4,5]
- Reductions in sBA and bilirubin are predictors of longer native liver survival in patients with PFIC who have had surgical biliary diversion to interrupt enterohepatic circulation.[6,7]
- Maralixibat (MRX) is a minimally absorbed ileal BA transporter inhibitor that prevents enterohepatic bile acid recirculation and is approved for the treatment of cholestatic pruritus in patients with PFIC ≥12 months of age in the US and for the treatment of PFIC in patients ≥3 months of age in the EU.[8,9]
- MARCH (NCT03905330) was a phase 3, randomized, double-blind, placebo-controlled, 26-week trial investigating the efficacy and safety of maralixibat in participants with PFIC across the broadest range of PFIC types studied to date.[10,11]
 - In MARCH, participants who received maralixibat achieved statistically significant improvements in pruritus, levels of sBAs and bilirubin, and growth.[11]
 - Significant and sustained responses were observed with up to 2 years of maralixibat treatment in MARCH-ON (NCT04185363), an open-label extension study for participants who completed the MARCH trial.[12,13]
- Improvements in pruritus and bilirubin in participants treated with maralixibat in MARCH/MARCH-ON were strongly correlated with reductions in total sBA.[11,14]

Objective

- To report the correlation between changes in sBA subspecies and changes in pruritus and direct bilirubin for participants with PFIC who received maralixibat in the MARCH/MARCH-ON trials.

Methods

- Change from Baseline (CFB) to Week 26 (averaged over the last 8-12 weeks) was calculated for all participants treated with maralixibat for 26 weeks in either MARCH or MARCH-ON.
- Spearman correlation coefficients were determined to evaluate the relationship between changes in sBA subspecies and changes in Itch-Reported Outcome (Observer) (ItchRO[Obs]) and direct bilirubin.
 - ItchRO(Obs) is a 0 to 4 scale, where 0 = no itch, 1 = mild, 2 = moderate, 3 = severe, and 4 = very severe.[15] A ≥1-point reduction in ItchRO(Obs) is considered clinically meaningful.
- Participants were stratified into responders (R) and nonresponders (NR) based on changes in ItchRO(Obs) and sBA after maralixibat treatment.
 - ItchRO(Obs) response was defined as a ≥1-point reduction in ItchRO(Obs) from Baseline to the average of the final 3 4-week periods in MARCH (Weeks 15-18, Weeks 19-22, and Weeks 23-26) or an average score of ≤1.[a]
 - sBA response was defined as an average sBA level of <102 μmol/L (if Baseline level was ≥102 μmol/L) or a ≥75% reduction from Baseline using the average from Weeks 18, 22, and 26.[a]

[a]Participants were defined as NR if the Baseline value or all 3 post-Baseline values were missing.

Results

- Sixty participants were included in the analysis, including the following PFIC types: BSEP (n=28), FIC1 (n=13), MDR3 (n=9), TJP2 (n=7), and MYO5B (n=3).
- Among the included participants, 88% (n=53) were receiving ursodeoxycholic acid (UDCA).

Decreases in Conjugated Primary BAs and Conjugated UDCA Were Correlated With Decreases in ItchRO(Obs) and Direct Bilirubin

Table 1. Serum Concentration of BA Subspecies and Correlation With Changes in ItchRO(Obs) and Direct Bilirubin in the All-PFIC Cohort

BA subspecies	Serum concentration of BA subspecies, μmol/L[a]		ItchRO(Obs) correlation coefficient (*P* value)	Direct bilirubin correlation coefficient (*P* value)
	Baseline	After MRX treatment[b]		
Total sBA	255 (180, 363)	105 (9, 199)	0.69 (<0.0001)	0.64 (<0.0001)
Conjugated primary BAs				
GCDCA	42 (27, 66)	27 (3, 46)	0.69 (<0.0001)	0.61 (<0.0001)
TCDCA	16 (9, 30)	8 (3, 16)	0.60 (<0.0001)	0.62 (<0.0001)
GCA	72 (34, 99)	16 (1, 62)	0.60 (<0.0001)	0.54 (<0.0001)
TCA	33 (17, 58)	12 (3, 29)	0.54 (0.0002)	0.50 (0.0008)
Unconjugated primary BAs				
CDCA	0.02 (0.02, 0.04)	0.2 (0.03, 0.6)	-0.33 (0.03)	-0.33 (0.03)
CA	0.02 (0.01, 0.03)	0.08 (0.03, 0.2)	-0.47 (0.03)	-0.35 (0.1)
Conjugated secondary BAs				
GDCA	0.06 (0.03, 0.2)	0.2 (0.05, 0.9)	-0.15 (0.3)	-0.33 (0.04)
TDCA	0.03 (0.01, 0.1)	0.07 (0.03, 0.2)	0.26 (0.2)	0.12 (0.6)
GLCA	0.02 (0.01, 0.04)	0.02 (0.01, 0.05)	-0.19 (0.6)	0.27 (0.4)
TLCA	0.02 (0.01, 0.03)	0.03 (0.02, 0.07)	-0.23 (0.6)	-0.41 (0.3)
GUDCA	46 (20, 120)	22 (1, 48)	0.40 (0.007)	0.44 (0.003)
TUDCA	6 (2, 19)	2 (1, 8)	0.41 (0.02)	0.64 (0.0001)
Unconjugated secondary BAs				
DCA	0.06 (0.01, 0.1)	0.2 (0.07, 0.5)	-0.50 (0.4)	-0.80 (0.1)
UDCA (from treatment)	0.6 (0.3, 3)	0.9 (0.5, 3)	0.27 (0.3)	0.31 (0.2)

[a]Data are median (Q1, Q3). [b]Average of Weeks 18, 22, and 26 of maralixibat treatment.

Table 2. Serum Composition of BA Subspecies by ItchRO(Obs) Response in the All-PFIC Cohort

BA subspecies	Average of Weeks 18, 22, and 26[a]			*P* value		
	PBO	NR	R	PBO vs NR	PBO vs R	NR vs R
Total unconjugated, μmol/L[b]	0.04 (0.0-0.6)	0.03 (0.02-0.7)	0.7 (0.1-4)	0.70	<0.001	<0.001
Unconjugated CA, μmol/L	0.02 (0.0-0.3)	0.02 (0.01-0.2)	0.1 (0.0-0.4)	0.84	<0.001	0.003
Unconjugated CDCA, μmol/L	0.02 (0.0-0.1)	0.02 (0.0-0.5)	0.5 (0.02-2)	0.74	<0.001	<0.001
Percentage unconjugated of total sBA[b]	0.01 (0.0-67)	0.02 (0.01-50)	17 (0.04-70)	0.91	<0.001	<0.001
Total secondary, μmol/L[c]	56 (0.03-373)	33 (0.04-230)	2 (0.0-113)	0.34	<0.001	0.032
Unconjugated UDCA (from treatment), μmol/L	0.09 (0.0-7)	0.2 (0.0-3)	0.2 (0.0-13)	0.71	0.10	0.42
Percentage secondary of total sBA[c]	31 (0.01-67)	20 (0.2-56)	21 (0.0-57)	0.083	0.16	0.55
G/T conjugation ratio[d]	3 (1-42)	6 (1-14)	11 (1-160)	0.071	<0.001	0.047
CA/CDCA ratio[e]	1 (0.0-3)	0.9 (0.4-1)	0.3 (0.0-1)	0.72	<0.001	<0.001

[a]Values shown are median (range). For ItchRO(Obs) response, the following number of participants were included in the analysis: placebo, n=31; NR, n=12; R, n=21. [b]Total unconjugated sBAs include CDCA, CA, DCA, and LCA. [c]Total secondary sBAs include GDCA, TDCA, GLCA, TLCA, GUDCA, TUDCA, DCA, and LCA. [d]G/T ratio is derived as the sum of GCDCA, GCA, GDCA, GLCA, and GUDCA divided by the sum of TCDCA, TCA, TDCA, TLCA, and TUDCA. [e]CA/CDCA ratio is derived as CA divided by CDCA.

Unconjugated Primary BAs Were Negatively Correlated With Markers of Disease in the All-PFIC Cohort

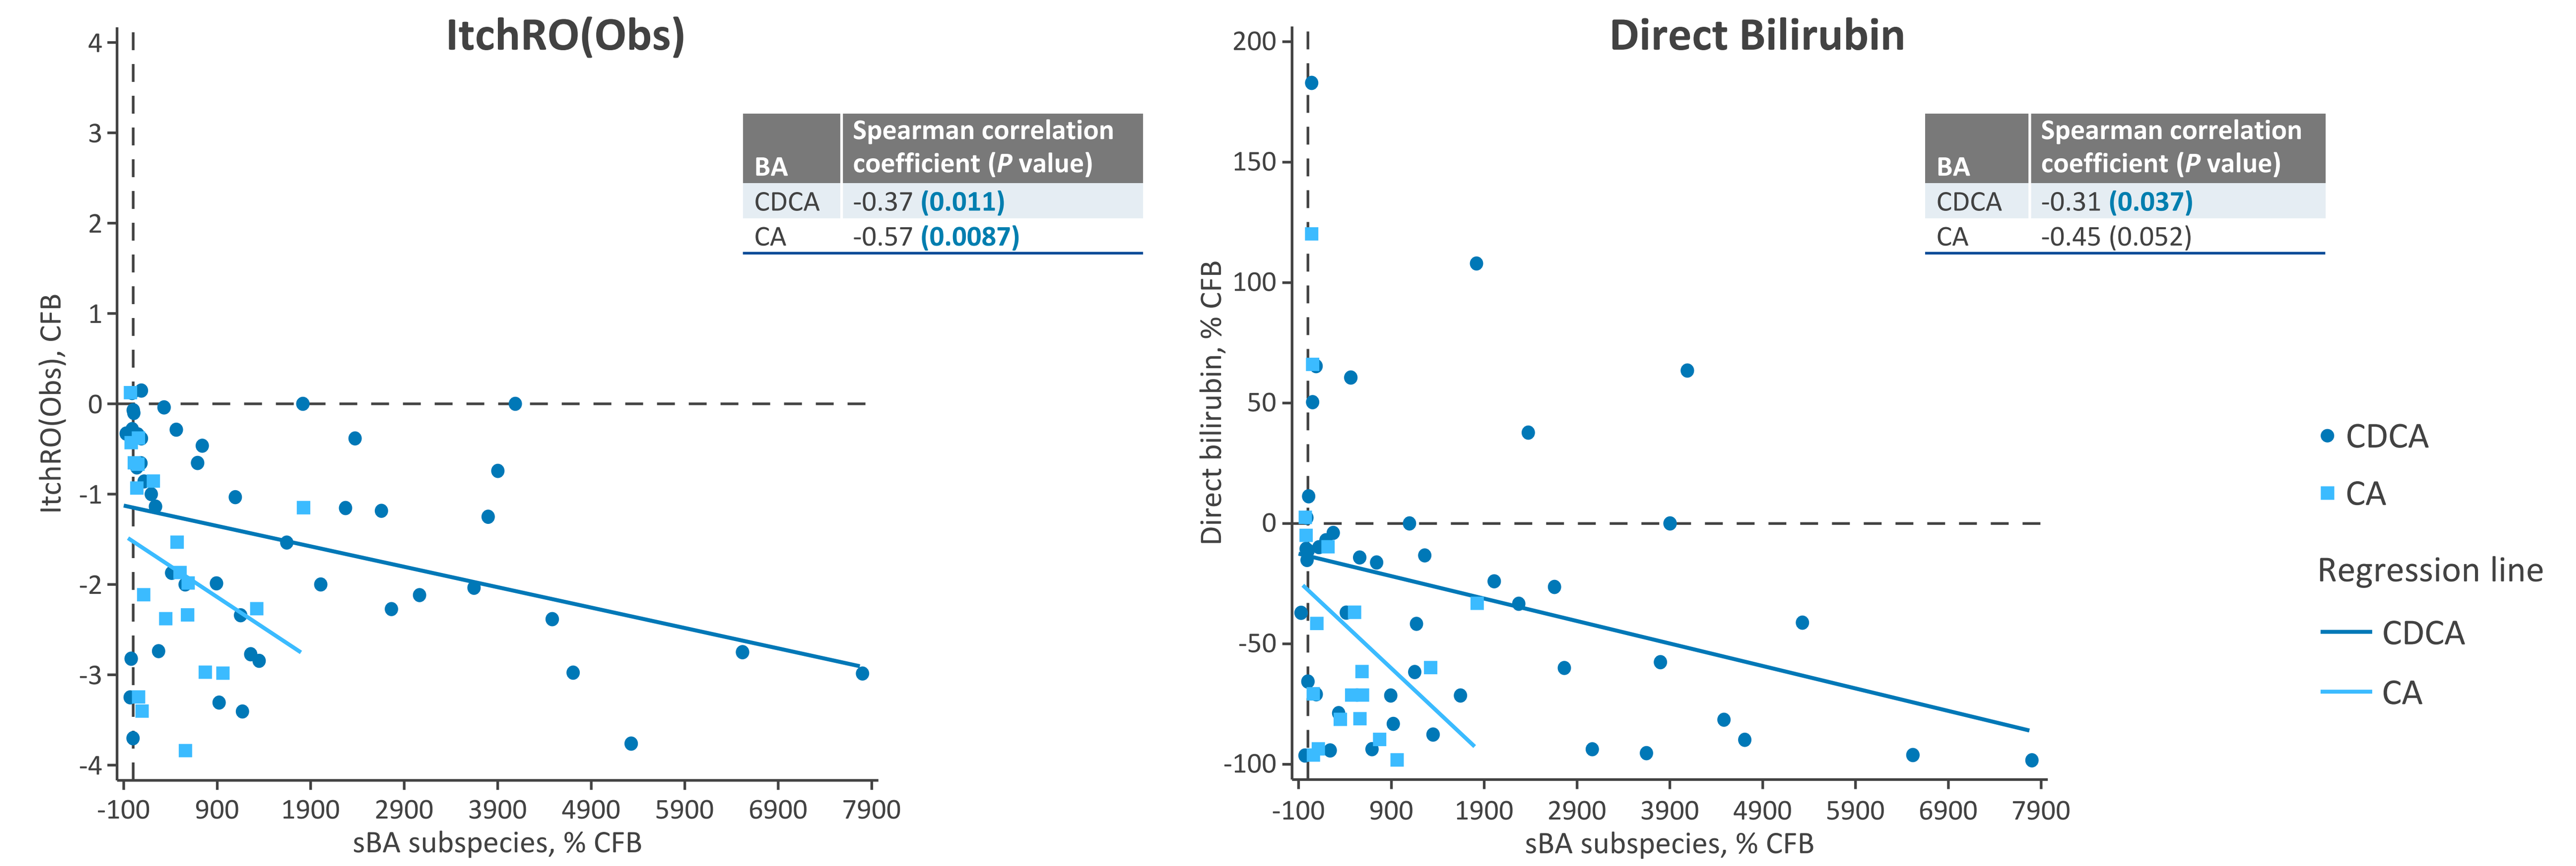
ItchRO(Obs)
ItchRO(Obs), CFB
sBA subspecies, % CFB
BA | Spearman correlation coefficient (P value)
CDCA | -0.37 (0.011)
CA | -0.57 (0.0087)
Direct Bilirubin
Direct bilirubin, % CFB
sBA subspecies, % CFB
BA | Spearman correlation coefficient (P value)
CDCA | -0.31 (0.037)
CA | -0.45 (0.052)
CDCA
CA
Regression line
CDCA
CA

Conjugated Primary BAs Were Positively Correlated With Markers of Disease in the All-PFIC Cohort

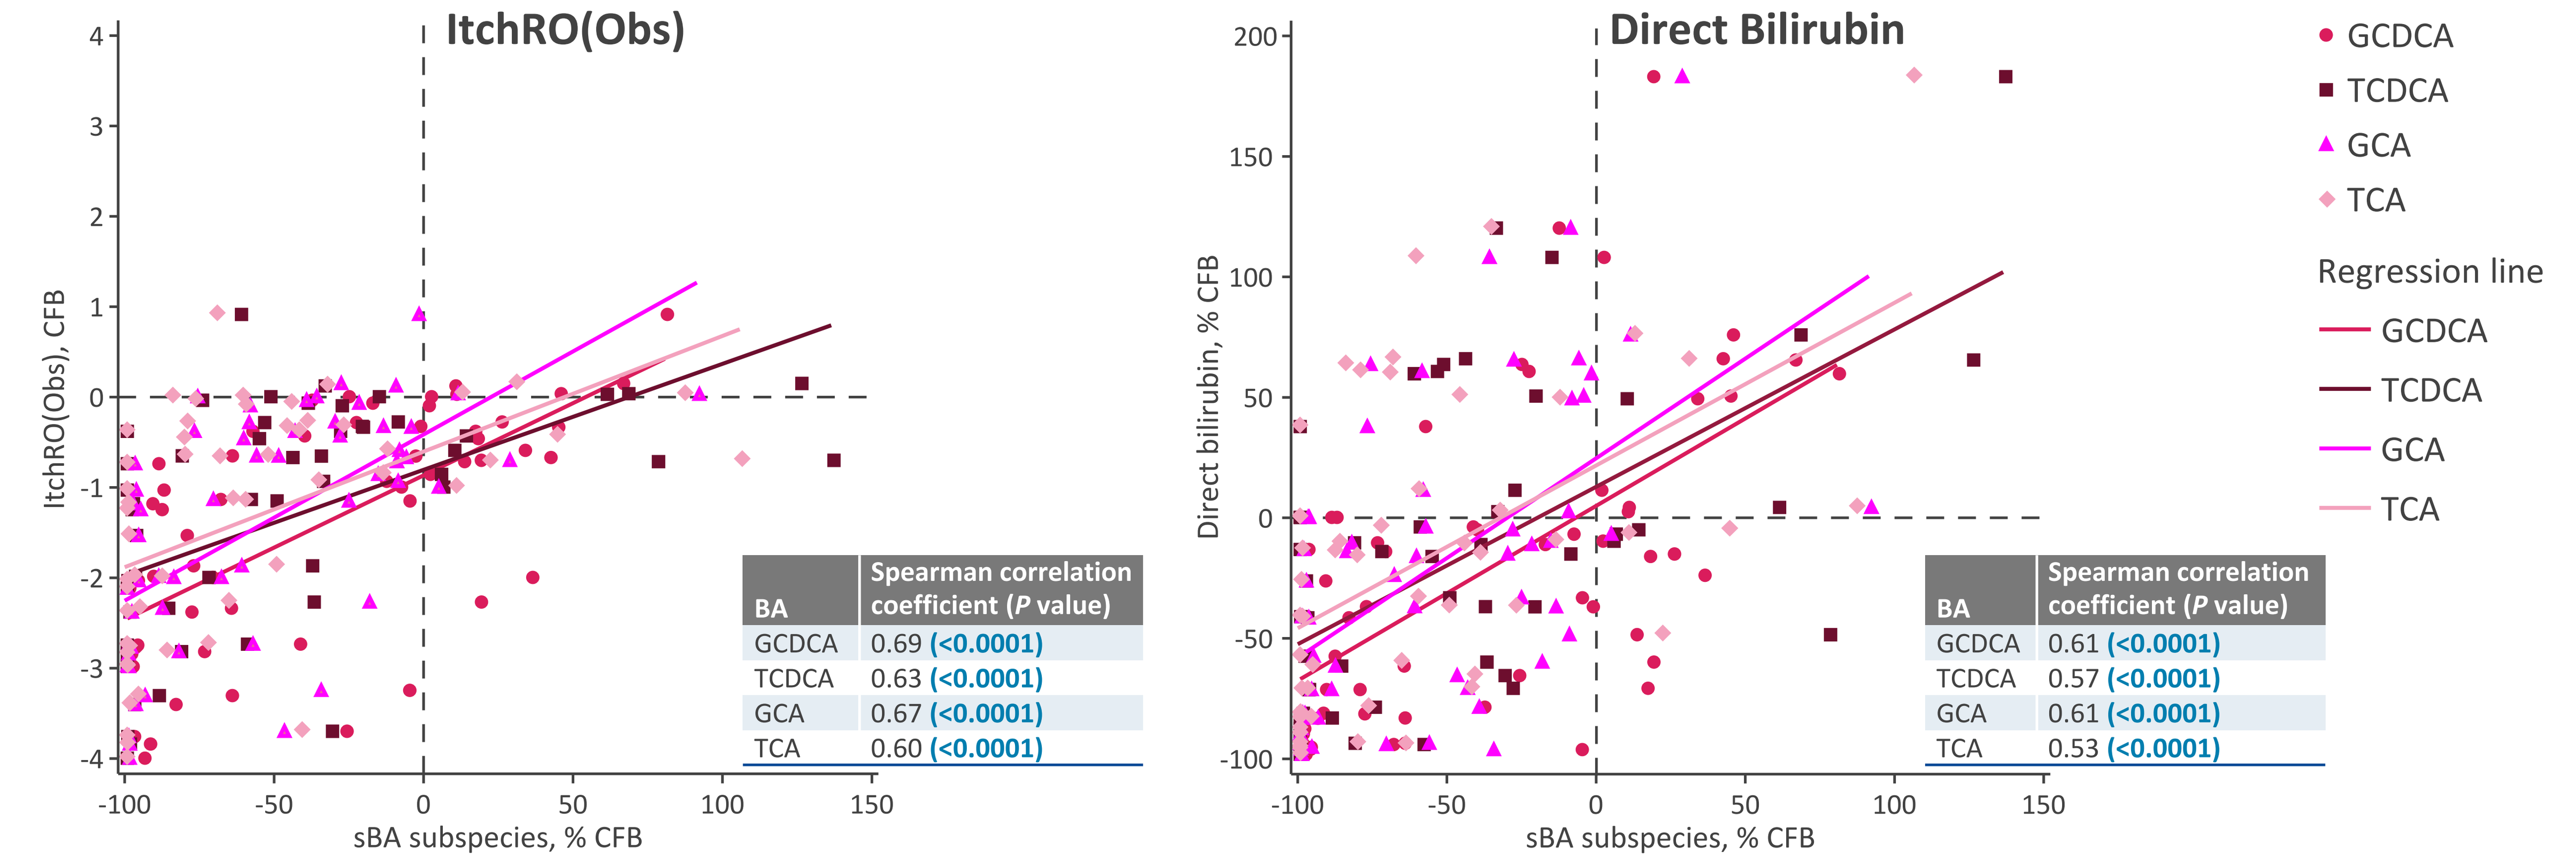
ItchRO(Obs)
ItchRO(Obs), CFB
sBA subspecies, % CFB
BA | Spearman correlation coefficient (P value)
GCDCA | 0.69 (<0.0001)
TCDCA | 0.63 (<0.0001)
GCA | 0.67 (<0.0001)
TCA | 0.60 (<0.0001)
Direct Bilirubin
Direct bilirubin, % CFB
sBA subspecies, % CFB
BA | Spearman correlation coefficient (P value)
GCDCA | 0.61 (<0.0001)
TCDCA | 0.57 (<0.0001)
GCA | 0.61 (<0.0001)
TCA | 0.53 (<0.0001)
GCDCA
TCDCA
GCA
TCA
Regression line
GCDCA
TCDCA
GCA
TCA

- Similar correlations were observed for participants in the BSEP cohort who received maralixibat (scan QR code to see data).
- For participants who received placebo, there were no significant correlations between BA subspecies and ItchRO(Obs) or direct bilirubin.

Conclusions

- These results expand on previous research demonstrating a correlation between sBA and pruritus and identify changes in BA subspecies associated with improvements in pruritus and direct bilirubin, including negative correlations with unconjugated primary BAs and positive correlations with conjugated primary BAs.
- The pattern of correlations indicates new BA synthesis in response to reduced BA levels in the liver, loss of FGF-19 mediated suppression, or increased bacterial action on BAs in the colon.
- In the All-PFIC and BSEP cohorts, serum composition of BA subspecies was significantly different between ItchRO(Obs) responders and the placebo group or ItchRO(Obs) nonresponders after treatment with maralixibat.
 - Similarly, sBA responders showed significant differences in BA composition compared with the placebo group or sBA nonresponders in the All-PFIC and BSEP cohorts (scan QR code to see data).
- These data help elucidate the pathophysiology of how maralixibat may improve both pruritus and underlying liver health in patients with PFIC.

Scan QR code for additional data

Abbreviations

BA, bile acid; BSEP, bile salt export pump; CA, cholic acid; CDCA, chenodeoxycholic acid; CFB, change from Baseline; DCA, deoxycholic acid; FGF19, fibroblast growth factor 19; FIC1, familial intrahepatic cholestasis-associated protein 1; GCA, glycocholic acid; GCDCA, glycochenodeoxycholic acid; GDCA, glycodeoxycholic acid; GLCA, glycolithocholic acid; GUDCA, glycoursodeoxycholic acid; G/T, glycine-to-taurine ratio; ItchRO(Obs), Itch-Reported Outcome (Observer); LCA, lithocholic acid; MDR3, multidrug resistance protein 3; MRX, maralixibat; MYO5B, myosin Vb; NR, nonresponder; PBO, placebo; PFIC, progressive familial intrahepatic cholestasis; R, responder; sBA, serum bile acid; TCA, taurocholic acid; TCDCA, taurochenodeoxycholic acid; TDCA, taurodeoxycholic acid; TJP2, tight junction protein 2; TLCA, taurolithocholic acid; TUDCA, tauroursodeoxycholic acid; UDCA, ursodeoxycholic acid.

Disclosures

HJV has nothing to declare. AGM is a consultant and has a sponsored research agreement with Mirum Pharmaceuticals, Inc. SPH is a hepatic safety adjudication committee member at Albireo and has received a research grant from Mirum Pharmaceuticals, Inc. DBM, TN, CK, JS, WG, and PV are employees of and shareholders in Mirum Pharmaceuticals, Inc. RJT is a consultant for Mirum Pharmaceuticals, Inc., Albireo, Generation Bio, Rectify Therapeutics, and Alnylam and a shareholder in Generation Bio and Rectify Therapeutics.

Acknowledgments

The authors would like to thank the clinical trial participants, their families, and the investigators for their participation in this study. Maralixibat is owned by Mirum Pharmaceuticals, Inc. This analysis was funded by Mirum Pharmaceuticals, Inc. Medical writing and editorial support for the development of this poster was provided by Precision AQ in Bethesda, Maryland, which was funded by Mirum Pharmaceuticals, Inc.

References

1. Kamath BM, et al. *Liver Int*. 2020;40:1812-1822. **2.** Rodrigo M, et al. *Biology (Basel)*. 2023;12:756. **3.** Patel SP, et al. *J Am Acad Dermatol*. 2019;81:1371-1378. **4.** Nomden M, et al. *Gastroenterology*. 2023;165(2):496-498.e1. **5.** Hanafi NI, et al. *Biomolecules*. 2018;8(4):159. **6.** van Wessel DBE, et al. *J Hepatol*. 2020;73:84-93. **7.** Bolia R, et al. *Expert Rev Gastroenterol Hepatol*. 2022;16:163-172. **8.** LIVMARLI® (maralixibat) [prescribing information]. Foster City, CA; Mirum Pharmaceuticals, Inc. Jul 2024. **9.** LIVMARLI® (maralixibat) [summary of product characteristics]. Amsterdam, Netherlands; Mirum Pharmaceuticals International B.V. Jul 2024. **10.** ClinicalTrials.gov identifier: NCT03905330. Updated December 11, 2023. Accessed September 16, 2024. https://clinicaltrials.gov/study/NCT03905330. **11.** Miethke AG, et al. *Lancet Gastroenterol Hepatol*. 2024;9:620-631. **12.** ClinicalTrials.gov identifier: NCT04185363. Updated May 29, 2024. Accessed September 12, 2024. https://www.clinicaltrials.gov/study/NCT04185363. **13.** Miethke A, et al. Oral presentation at AASLD 2023. **14.** D'Antiga L, et al. Presented at EASL 2024. **15.** Kamath BM, et al. *Hepatol Commun*. 2020;4:1012-1018.

Presented at American Association for the Study of Liver Diseases (AASLD) The Liver Meeting®; November 15-19, 2024; San Diego, California